P2107

Characterizing Mild and Moderate Hemophilia A Patients in the Real World: A Patient-Centric Approach

Michelle Witkop,[1*] Tao Xu,[2] Gillian Hanson,[3] Richard Ofori-Asenso,[4] Richard H. Ko,[5] Emily Cibelli,[3] Martynas Aizenas,[2] Mark W. Skinner,[6,7] Fabian Sanabria,[2] Amy Shapiro[8]

*presenting author

Summary

We utilized a novel methodology to conduct cohort studies for people with hemophilia A

Medical records were available over a median of 6.3 years per person

6.3 years

The cohort building and data collection methodology is proven to be valid for hemophilia A

Preliminary results suggest congruence with the Centers for Disease Control and Prevention (CDC) Community Counts

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASH® and the author of this poster. Download this presentation: https://bit.ly/31ddQIz

INTERACTIVE

[1]National Hemophilia Foundation, NY, USA; [2]F. Hoffmann-La Roche Ltd, Basel, Switzerland; [3]PicnicHealth, San Francisco, CA, USA; [4]Roche Products Ltd, Welwyn Garden City, UK; [5]Genentech, Inc., South San Francisco, CA, USA; [6]McMaster University, Hamilton, ON, Canada; [7]Institute for Policy Advancement Ltd, Washington DC, USA; [8]Indiana Hemophilia and Thrombosis Center, Indianapolis, IN, USA

Background

- People with mild or moderate hemophilia A (HA) account for 40–52% of all people with HA (PwHA) but are under-represented in scientific research.[1,2]
- To address this gap, we utilized a novel, patient-centric approach developed by PicnicHealth to create a longitudinal healthcare database from people with mild or moderate HA in the United States (US).
- This study integrates data collected during routine clinical care from various providers and sites along with patient-reported outcomes (PROs) on an online record management platform to provide an in-depth characterization of patients' disease journey.

Methods

- PicnicHealth HA study recruitment commenced in June 2020 following ethics approval. Consented eligible PwHA had a documented HA diagnosis.
- Severity status was confirmed based on physician-reported severity in provider notes or reported baseline factor (F)VIII level (>5–40% mild, 1–5% moderate); the lowest FVIII value was used if notes and baseline levels were inconclusive.
- Data elements from structured text (e.g. medication lists), as well as disease-specific elements from narrative text, were captured from patients' electronic health records and linked to self-reported data (**Figure 1**). PROs, including bleeds, treatment, physical and social activity, and pain, were collected biweekly via online questionnaire for a subset of PwHA.
- Quality control was assessed via inter-abstractor agreement on outputs with physician review. Descriptive analyses were performed to summarize cohort characteristics and demonstrate the breadth and completeness of the data.
- Cohort characteristics were compared with data on people with mild or moderate HA from the Centers for Disease Control and Prevention (CDC) Community Counts,[3] a public health monitoring program for people with bleeding disorders.
- For details about the data collection process, please visit our talk on **Monday, December 13, 2021, at 11:45 am in Session 322: A Novel Methodology for Building Longitudinal, Patient-Centric Real-World Datasets in Hemophilia A** (MW Skinner, et al. Abstract #594).

Figure 1. Data collection by PicnicHealth

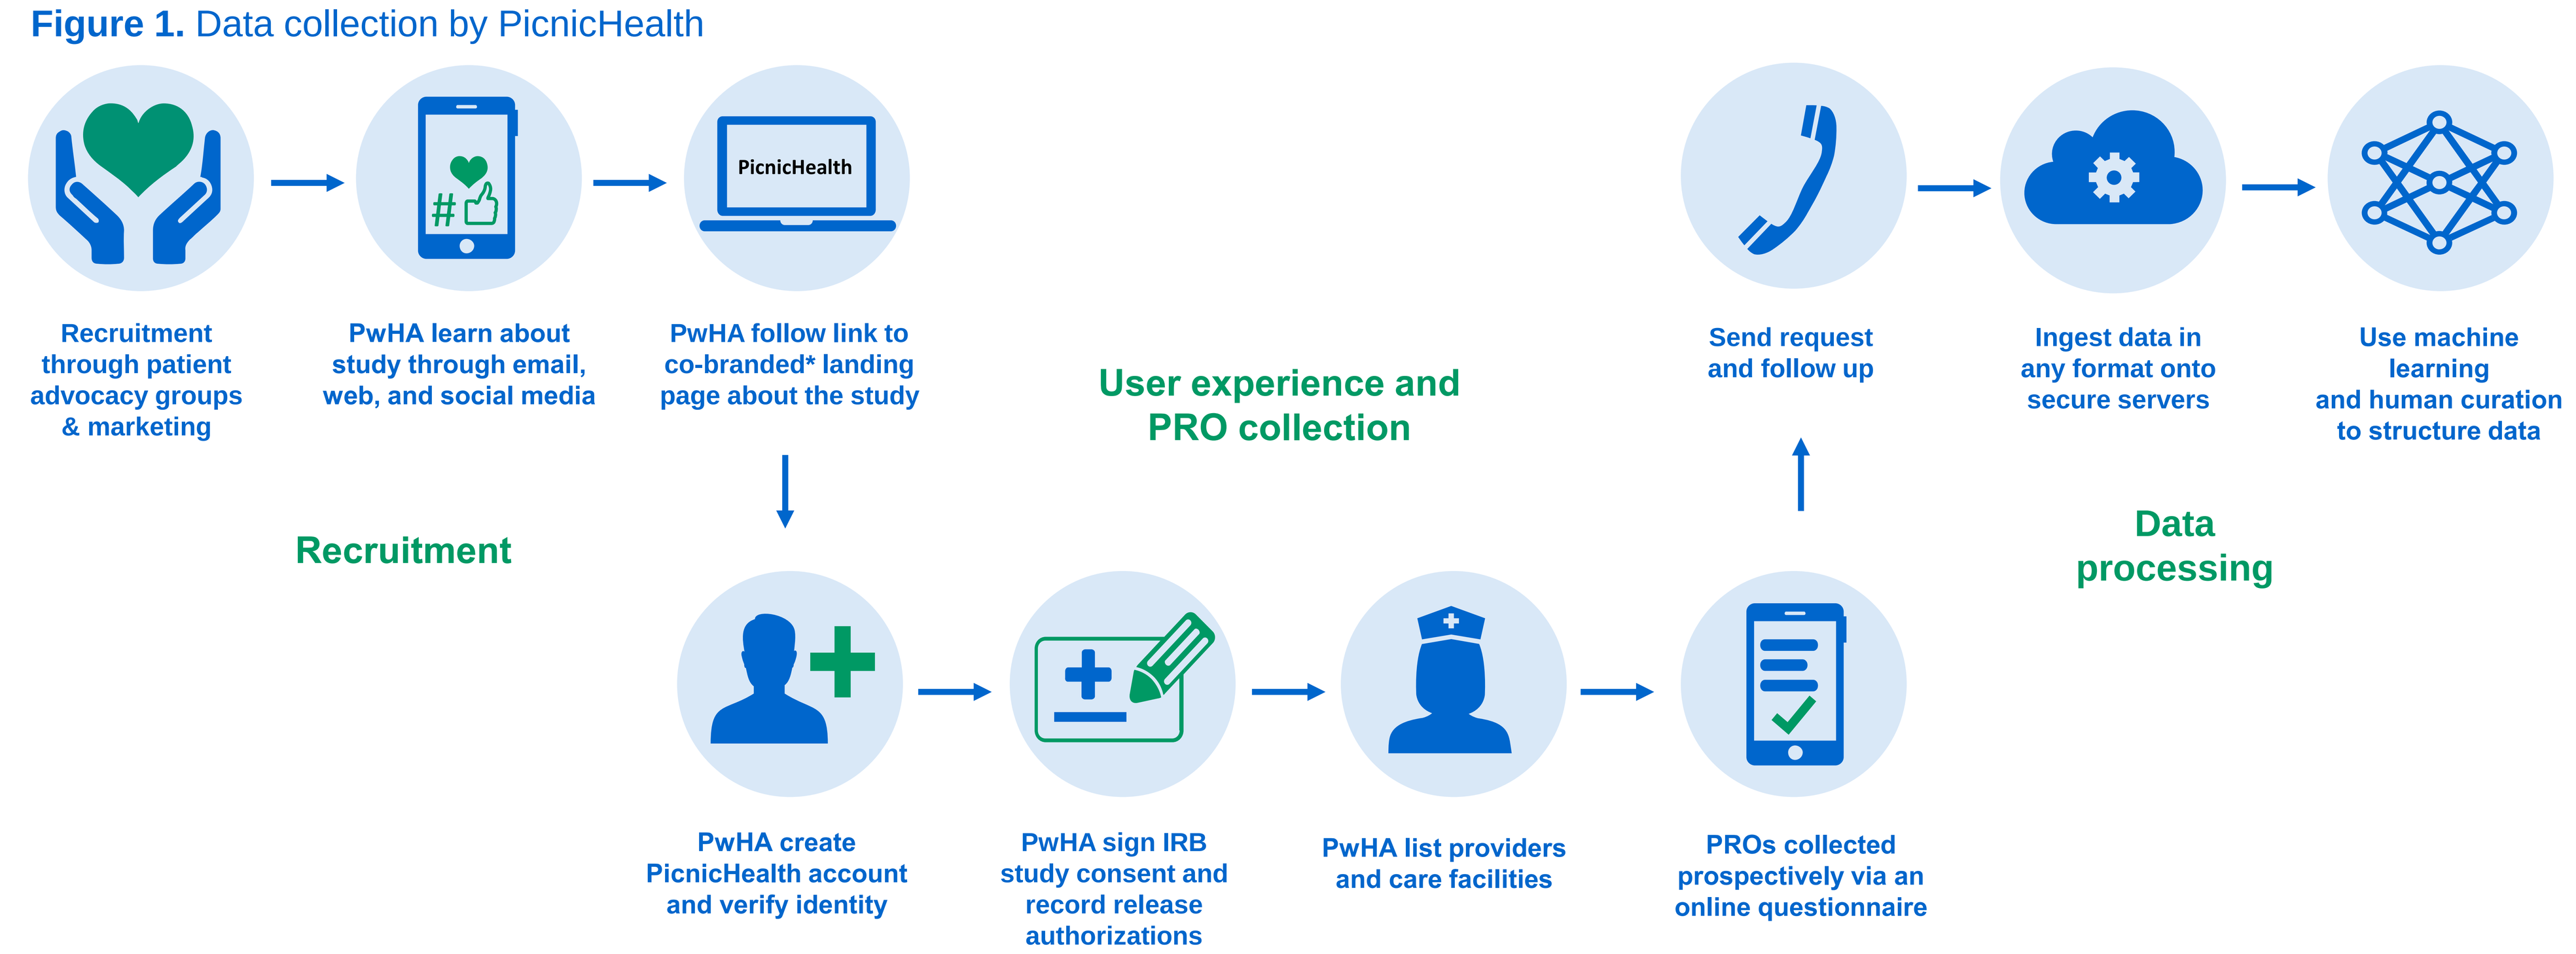

Recruitment through patient advocacy groups & marketing
PwHA learn about study through email, web, and social media
PicnicHealth
PwHA follow link to co-branded* landing page about the study
Recruitment
PwHA create PicnicHealth account and verify identity
PwHA sign IRB study consent and record release authorizations
PwHA list providers and care facilities
PROs collected prospectively via an online questionnaire
User experience and PRO collection
Send request and follow up
Ingest data in any format onto secure servers
Use machine learning and human curation to structure data
Data processing

*Study conducted as part of a strategic partnership with F. Hoffmann-La Roche, Ltd.
IRB, institutional review board

Results: data collection

- As of June 2021, 143 PwHA from 32 states in the US had signed up to the study (**Figure 2**), of whom 104 met eligibility criteria for enrollment.
- Of these, 65 (62.5%) had mild HA, including 23 females, and 39 (37.5%) had moderate HA.
- Medical records were available over a median (interquartile range [IQR]) of 6.3 (3.7–11.1) years from 16 (8–28) healthcare providers of six (3–10) care sites for the cohort before the enrollment dates.
- In total, data were collected from 588 care sites and 1963 providers, including 234 hematology and 1729 non-hematology providers, with specialties including radiology, family medicine, pediatrics and psychiatry.
- PROs were collected prospectively in 25 PwHA starting from February 25, 2021, with the latest data collected in June 2021
 - On average, the response rate was 90.3% (150/166 requests)
 - Of the 25, 10 (40.0%) responded to all questions at the expected time, nine (36.0%) missed one response, and six (24.0%) had more than one missing.

Figure 2. Geographic distribution of the PwHA

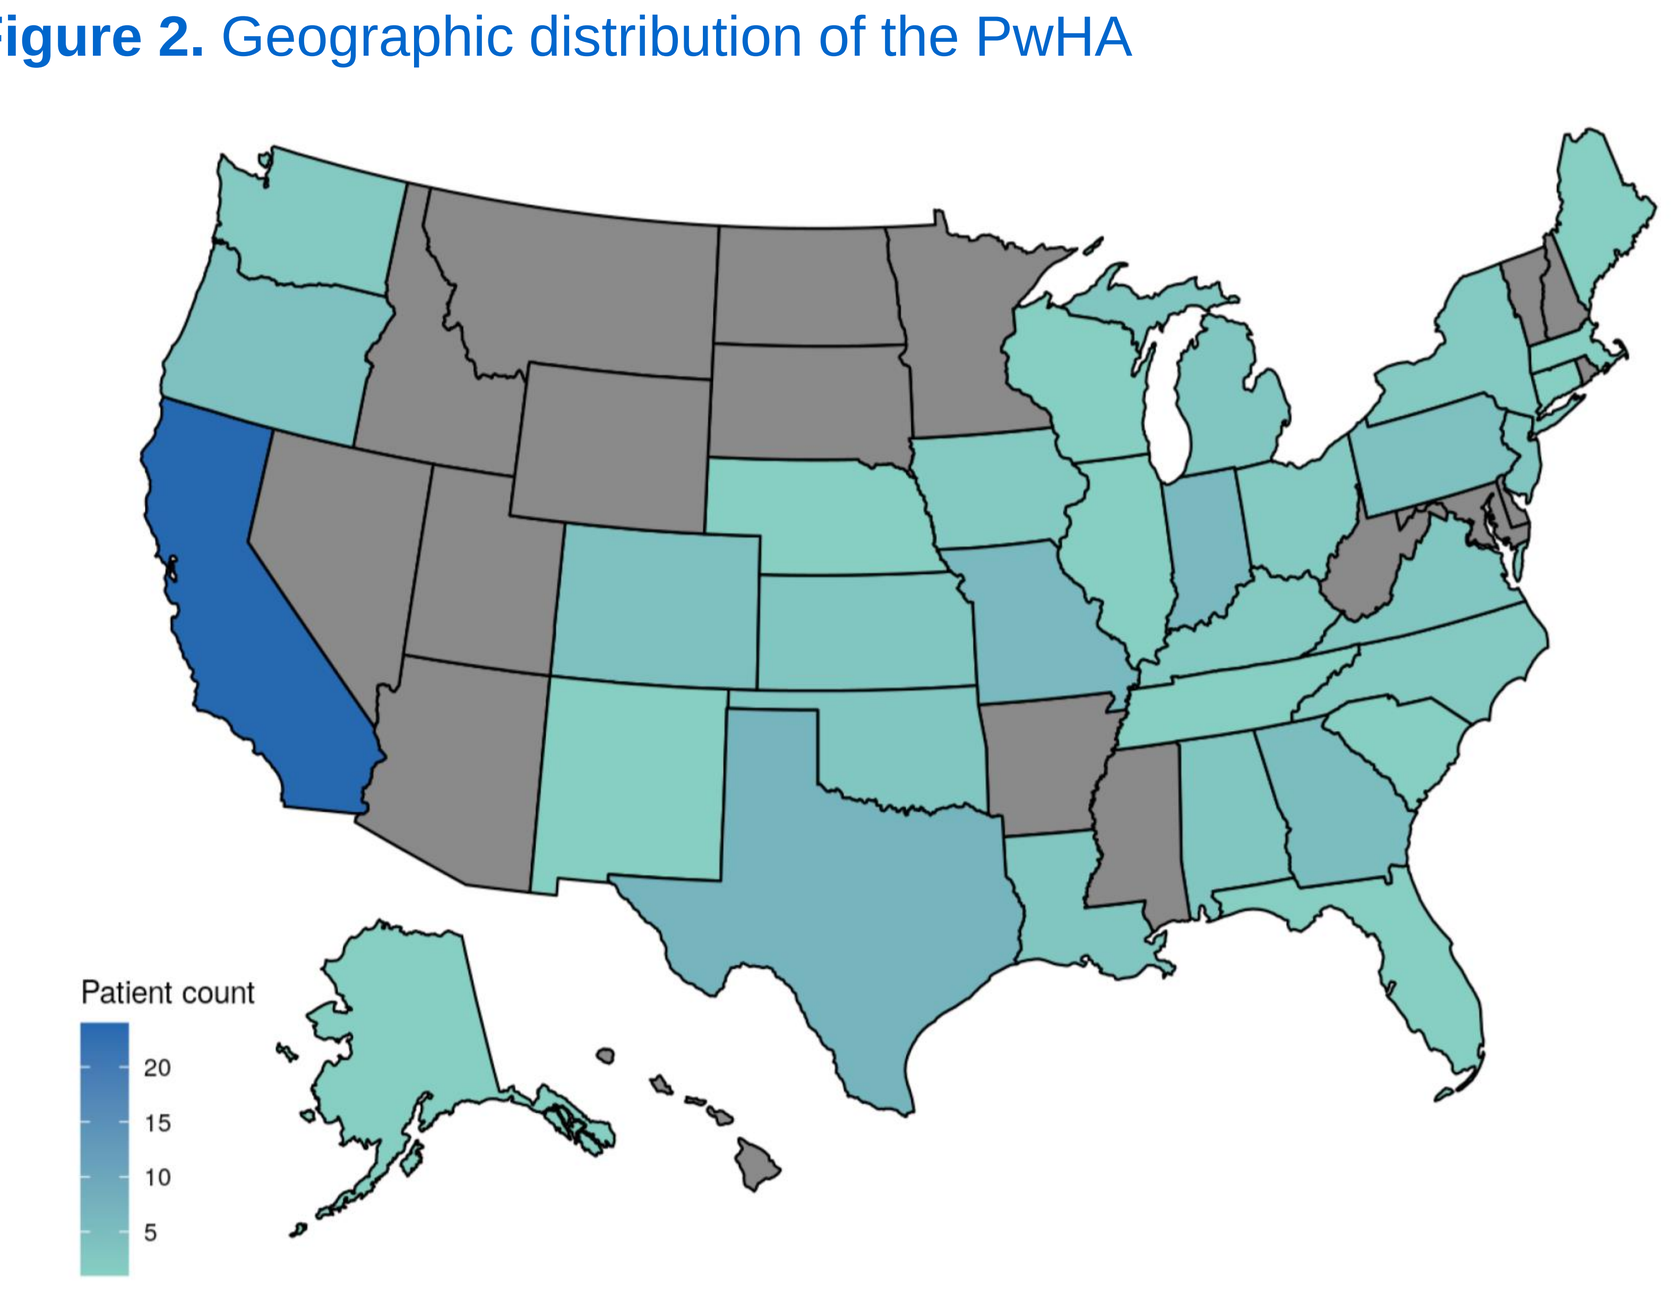

Patient count
20
15
10
5

Results: cohort characteristics

- The median (interquartile range [IQR]) age at enrollment was 29 (22–42) years.
- Age at first diagnosis was available for 55 (52.9%) PwHA; the median (IQR) was 3 (0–13) years
 - Males were diagnosed 10 years younger than females (males, 2 [0–9] years; females, 12 [5–29] years)
 - People with moderate HA were diagnosed at a median (IQR) age of 1 (0–6) years; for people with mild HA, diagnosis was at 5 (0–16) years.
- With regard to disease severity and ethnicity distributions, the cohort was comparable with the CDC's Community Counts (**Table 1**)
 - However, the present study recruited a higher proportion of female PwHA, which may be due to increasing awareness of PwHA with menstruating potential, and a lower proportion of the pediatric population.

Table 1. Cohort characteristics in comparison with CDC Community Counts

	PicnicHealth HA cohort			CDC Community Counts		
	All N=104	Males n=81 (77.9%)	Females n=23 (22.1%)	All N=4905	Males n=4383 (89.4%)	Females n=522 (10.6%)
Severity of hemophilia,* n (%)						
Moderate	39 (37.5)	39 (48)	0 (0)	1825 (37.2)	1797 (41)	28 (5.4)
Mild	65 (62.5)	42 (52)	23 (100)	3080 (62.8)	2586 (59)	494 (94.6)
Age group (at enrollment), n (%)						
<2 years	0 (0)	0 (0)	0 (0)	220 (4.5)	209 (4.8)	11(2.1)
2–10 years	10 (9.6)	7 (8.6)	3 (13.0)	1045 (21.3)	971 (22.2)	74 (14.2)
11–19 years	11 (10.6)	10 (12.3)	1 (4.3)	1109 (22.6)	1002 (22.9)	107 (20.5)
20–44 years	60 (57.7)	46 (56.8)	14 (60.9)	1425 (29.1)	1227 (28.0)	198 (37.9)
45–64 years	19 (18.3)	14 (17.3)	5 (21.7)	784 (16.0)	682 (15.6)	102 (19.5)
65+ years	4 (3.8)	4 (4.9)	0 (0)	322 (6.6)	292 (6.7)	30 (5.8)
	(n=55)	(n=48)	(n=7)			
Median age at first hemophilia diagnosis (IQR), years	3 (0–13)	2 (0–9)	12 (5–29)	N/A	N/A	N/A
Ethnicity, n (%)						
Hispanic, Latino/a, or Spanish origin	9 (8.7)	7 (8.6)	2 (8.7)	933 (19.0)	843 (19.2)	90 (17.2)
Not Hispanic, Latino/a, or Spanish origin, or unknown	95 (93.1)	74 (91.4)	21 (91.3)	3972 (81.0)	3540 (80.8)	432 (82.8)
Race, n (%)						
More than one race or unknown	38 (36.5)	31 (38.3)	7 (30.4)	252 (5.1)	220 (5.0)	32 (6.1)
White	61 (58.7)	46 (56.8)	15 (65.2)	4017 (81.9)	3595 (82.0)	422 (80.8)
Black or African American	4 (3.8)	3 (3.7)	1 (4.3)	449 (9.2)	407 (9.3)	42 (8.1)
Other races	1 (1.0)	1 (1.2)	0 (0.0)	187 (3.8)	161 (3.7)	26 (5.0)
BMI status (age >19 years),† n (%)	(N=83)	(N=64)	(N=19)	(N=2531)	(N=2201)	(N=330)
Underweight (<18.5 kg/m²)	2 (2.4)	1 (1.6)	1 (5.3)	28 (1.1)	23 (1.1)	5 (1.5)
Normal (18.5–24.9 kg/m²)	13 (15.7)	13 (20.3)	0 (0)	781 (30.9)	670 (30.4)	111 (33.6)
Overweight (25.0–29.9 kg/m²)	16 (19.3)	12 (18.8)	4 (21.1)	659 (26.0)	597 (27.1)	62 (18.8)
Obese (≥30.0 kg/m²)	28 (33.7)	19 (29.7)	9 (47.4)	877 (34.7)	757 (34.4)	120 (36.4)
Unreported	24 (28.9)	19 (29.7)	5 (26.3)	186 (7.4)	154 (7.0)	32 (9.7)

CDC Community Counts data are from the Hemophilia Treatment Center Population Profile Patient Characteristics. Accessible at: https://www.cdc.gov/ncbddd/hemophilia/communitycounts/data-community-counts.html. *One symptomatic person with HA in the PicnicHealth cohort had FVIII levels >40%; for the purposes of this study, they are included in the mild HA cohort; †For the PicnicHealth cohort, weight and height information within one year before enrollment was used in the BMI calculation. Percentages were calculated within the age group.
BMI, body mass index; IQR, interquartile range; N/A, not available.

- In the year before enrollment, 83 PwHA had outpatient visits, with a median of three visits for the cohort; 20 PwHA had inpatient admissions (**Table 2**).
- The inpatient visit count was slightly higher in this cohort than CDC Community Counts, where 7.7% had 1 visit over the past year before enrollment, and 1.9% had 2–3 visits. Inpatient admission and outpatient visit frequencies in the year before enrollment were lowest in the 20–44 years age group (median [IQR] 1 [1–3]) (**Figure 3**).

Table 2. Inpatient admissions and outpatient visits of the cohort

	PicnicHealth HA cohort		
	All N=104	Males n=81 (77.9%)	Females n=23 (22.1%)
Inpatient admission count,* n (%)			
0 or unreported	84 (80.8)	67 (82.7)	17 (73.9)
1	12 (11.5)	9 (11.1)	3 (13.0)
2–3	6 (5.8)	4 (4.9)	2 (8.7)
>3	2 (1.9)	1 (1.2)	1 (4.3)
Outpatient visit count,* n (%)			
0 or unreported	21 (20.2)	16 (19.7)	5 (21.7)
1	22 (21.2)	18 (22.2)	4 (17.4)
2–3	23 (22.1)	18 (22.2)	5 (21.7)
>3	38 (36.5)	29 (25.8)	9 (39.1)
	CDC Community Counts		
	All N=4905	Males n=4383 (89.4%)	Females n=522 (10.6%)
Inpatient admission count,* n (%)			
0 or unreported	4418 (90.1)	3924 (89.5)	494 (94.7)
1	378 (7.7)	358 (8.2)	20 (3.8)
2–3	93 (1.9)	88 (2.0)	N/A†
>3	N/A†	N/A†	N/A†

*Data from one year before enrollment; †CDC Community Counts suppressed the categories with low counts, which are marked as N/A; all percentages were calculated based on the full cohort.

Figure 3. Median number of clinical (including inpatient and outpatient) visits by age group

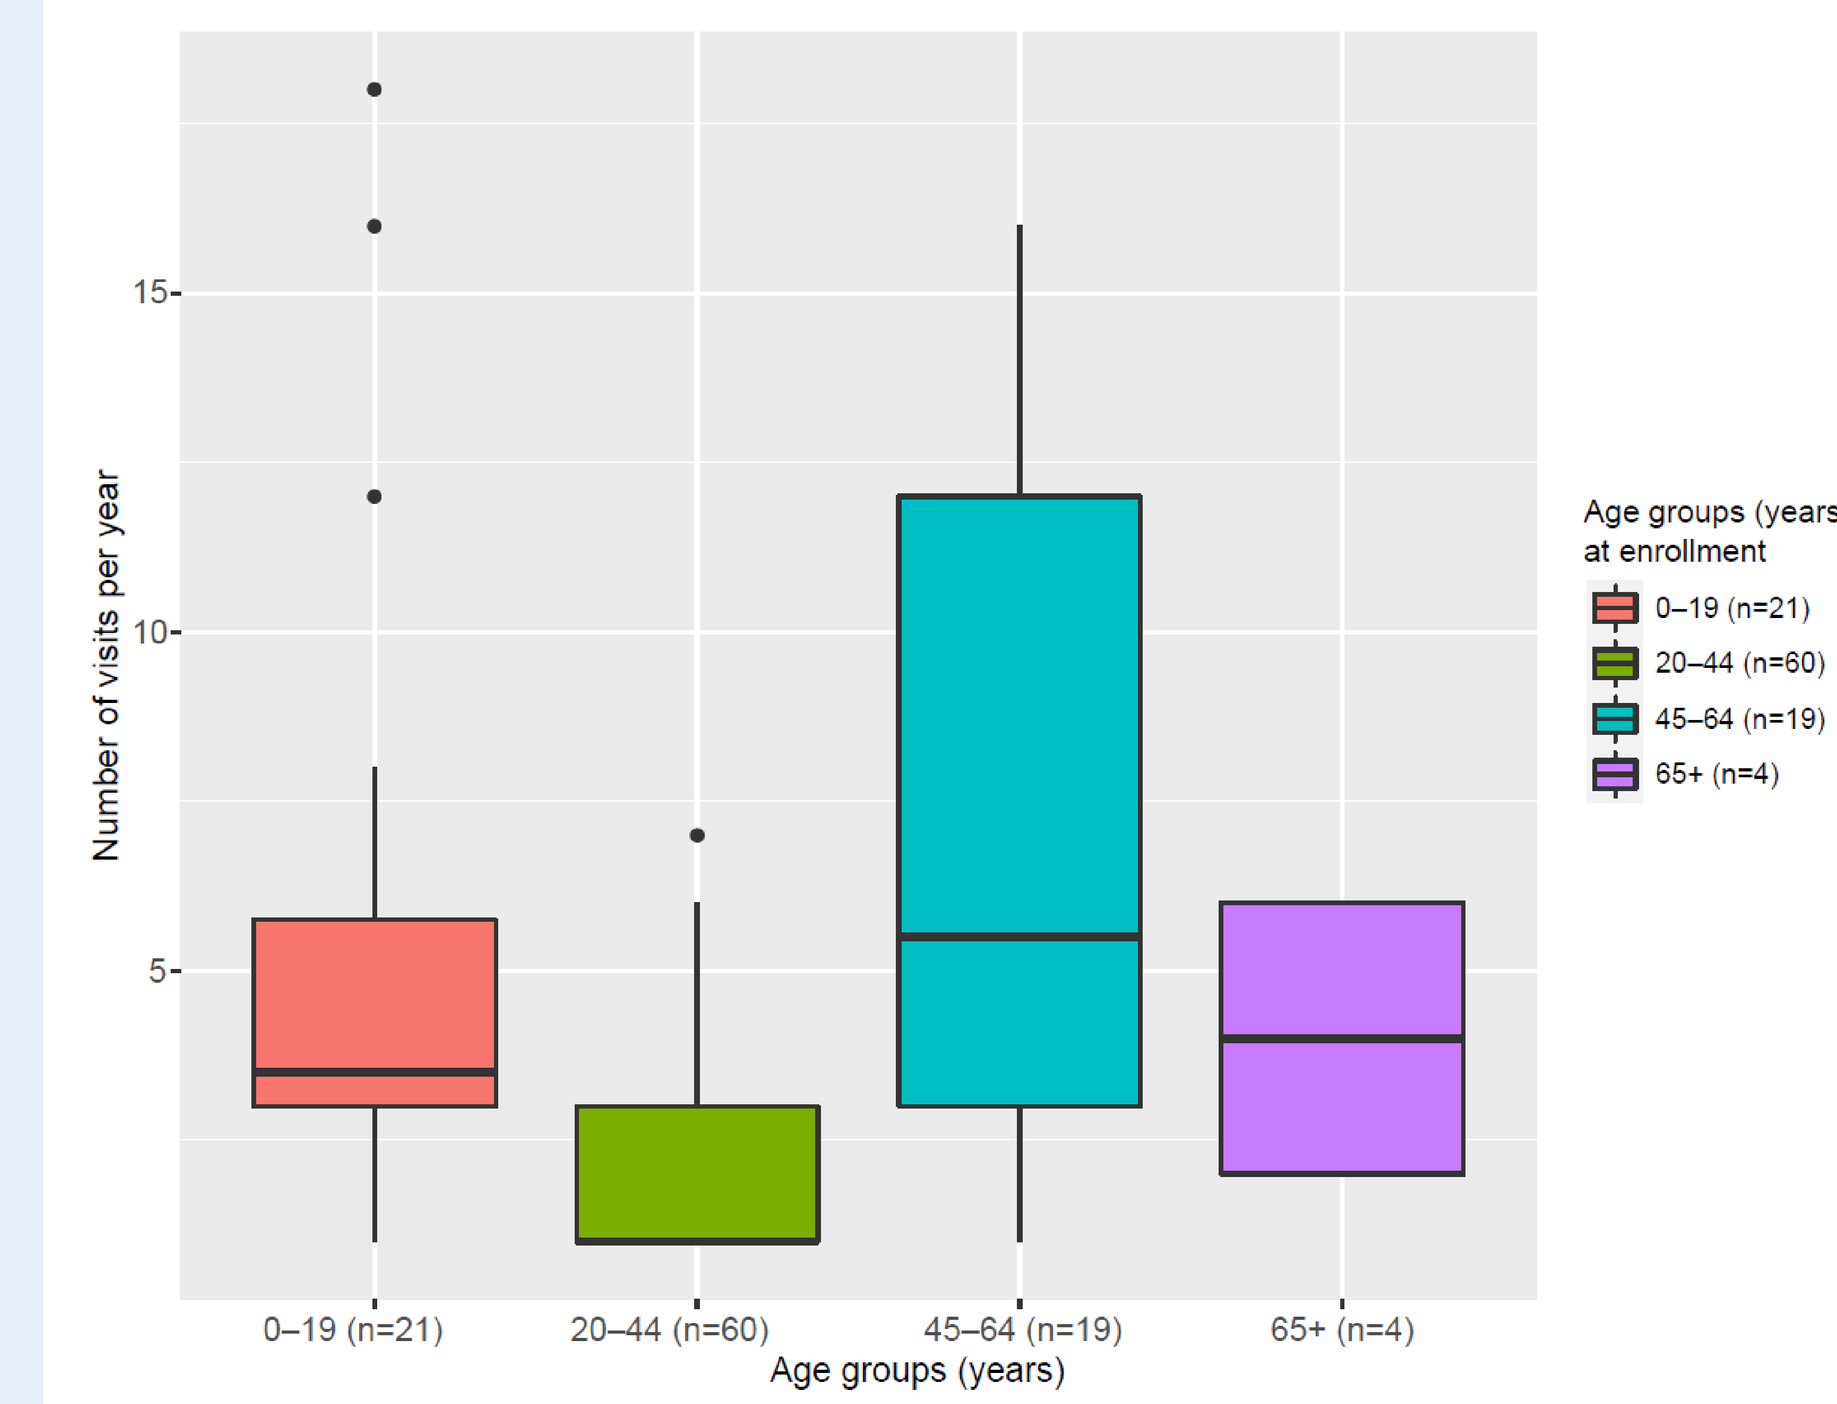

Number of visits per year
15
10
5
0–19 (n=21)
20–44 (n=60)
45–64 (n=19)
65+ (n=4)
Age groups (years)
Age groups (years) at enrollment
0–19 (n=21)
20–44 (n=60)
45–64 (n=19)
65+ (n=4)

Boxes represent the IQR of the number of clinical visits for each age group, with the median indicated by the thick black line. Whiskers represent the range, and outliers (outside 1.5 times the IQR) are indicated by dots.

Conclusions

- The patient-centric data collection methods implemented in this study provide a new approach to building cohorts for observational studies for PwHA.
- PicnicHealth combines routine clinical care data and PROs, and preliminary results suggest congruence when compared with an existing dataset. Participants have access to all records collected, organized in a medical timeline and shareable with care providers, potentially alleviating the burden of care coordination for the patient.
- The result is a patient-centric approach to data that benefits participants while providing needed data on groups traditionally under-represented in real-world evidence and traditional PwHA cohorts.

Presented at the 63rd ASH Annual Meeting and Exposition | 11–14 December 2021

References

1. Michele DM, et al. Haemophilia 2014;20:e136–43.
2. Benson G, et al. Blood Transfus 2018;16:535–44.
3. CDC Community Counts data are from the Hemophilia Treatment Center Population Profile Patient Characteristics. Available at: https://www.cdc.gov/ncbddd/hemophilia/communitycounts/data-community-counts.html. Accessed November 2021.

Acknowledgments

This study was sponsored by F. Hoffmann-La Roche Ltd and Genentech, Inc. Third party editorial assistance, under the direction of the authors, was provided by Phoebe Tate, MSc, of Ashfield MedComms, an Ashfield Health Company, and was funded by F. Hoffmann-La Roche Ltd.

Disclosures

MW: Current employment with National Hemophilia Foundation, consultancy role at Roche Advisory Panel. **TX, ROA, MA, FS**: current employment with Roche/Genentech and stock or other ownership in Roche. **RHK**: current employment with Roche/Genentech, stock or other ownership in Roche and other privately held companies. **GH, EC**: current employment with PicnicHealth and holder of stock options in PicnicHealth. **MWS**: current employment with Institute for Policy Advancement Ltd; consultancy role at National Hemophilia Foundation, current holder of stocks in IPA Ltd; research funding from BioMarin, Freeline, Roche, Takeda, Uniqure, honoraria from BioMarin, Takeda, Bayer, Pfizer (DMC), Roche, Sanofi, Spark (DMC); Membership on the Board of Directors or advisory committees of Bayer, ICER, WFH USA, BCBS MAP. **ADS**: current employment at Indiana Hemophilia & Thrombosis Center, Inc; consultancy role at Sangamo Therapeutics, ProMetic Life Sciences and Sigilon; research funding from ProMetic Life Sciences, Sigilon, Bioverativ/Sanofi, Genentech, Kedrion Biopharma, Novo Nordisk, Pfizer, Takeda, and Freeline; honoraria from Sigilon, Novo Nordisk, Pfizer, Genentech, and Catalyst Biosciences; membership on the Board of Directors or advisory committees of Sigilon, Novo Nordisk, Genentech, and Sanofi; Speakers Bureau at Novo Nordisk and Genentech.