PB0671

Analysis of *F8* genotypes and pharmacodynamic and safety biomarkers in people with moderate or mild hemophilia A receiving emicizumab ▼ in the HAVEN 6 trial

Anna Kiialainen,[1] Claude Negrier,[2] Steven W. Pipe,[3] Benjamin M. Beckermann,[1] Olivier Catalani,[1] Christophe Schmitt,[1] Cédric Hermans,[4] Johannes Oldenburg,[5] Michaela Lehle[1]

Summary

HAVEN 6 is a Phase III, single-arm, open-label trial exploring the use of emicizumab prophylaxis in people with moderate or mild hemophilia A (HA) without factor VIII inhibitors

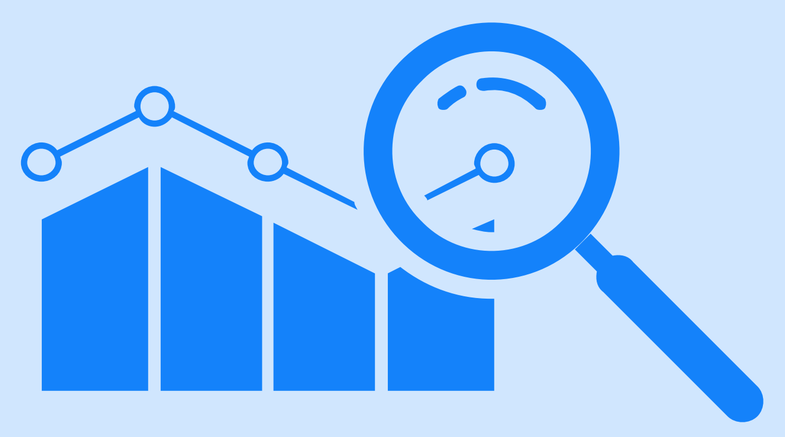

This exploratory analysis investigated *F8* genotypes, pharmacodynamics and exploratory safety biomarkers in HAVEN 6 participants

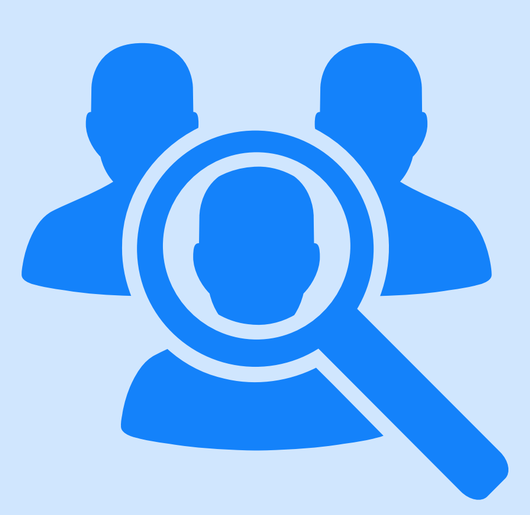

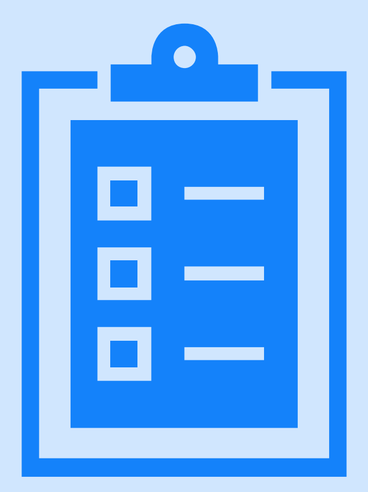

Emicizumab improved coagulation parameters without affecting endogenous factor VIII or safety biomarker levels

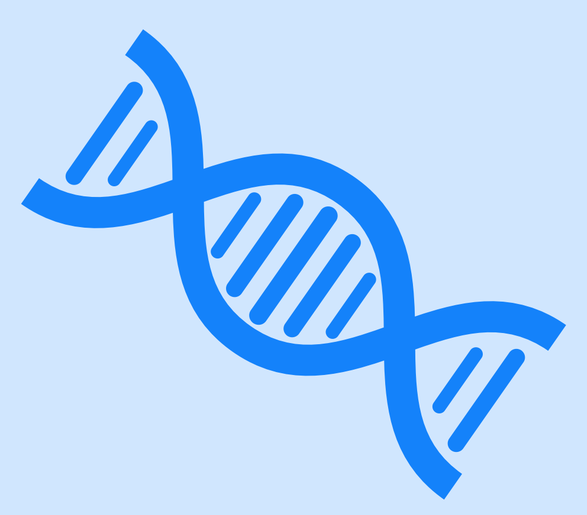

The identified *F8* mutations were consistent with mutations expected in people with moderate or mild HA

[1]*F. Hoffmann-La Roche Ltd, Basel, Switzerland;*
[2]*Claude Bernard Lyon 1 University, Lyon, France;*
[3]*University of Michigan, Ann Arbor, MI, USA;*
[4]*Cliniques Universitaires Saint-Luc, Brussels, Belgium;*
[5]*University of Bonn, Bonn, Germany.*

Background

- Emicizumab is a bispecific, monoclonal antibody that substitutes for the function of missing activated factor VIII (FVIII) in people with hemophilia A (HA), by bridging factor X and activated factor IX, thereby improving hemostasis.[1]
- In HAVEN 6 (NCT04158648), a Phase III, multicenter, single-arm, open-label trial in people with moderate or mild HA without FVIII inhibitors, emicizumab demonstrated a favorable safety and efficacy profile when given weekly (QW), every 2 weeks (Q2W) or every 4 weeks (Q4W).[2]
- Here, we present pharmacodynamics (PD) of emicizumab; an *F8* genetic analysis; and exploratory safety biomarkers in people with moderate or mild HA receiving emicizumab in the HAVEN 6 trial.

The HAVEN 6 trial includes people of all ages with moderate or mild HA without FVIII inhibitors

- PD analyses:
 - FVIII-like activity was measured using a chromogenic assay with human derived factors, as outlined by Schmitt *et al.*[3] It cannot be compared with, or interpreted as equivalent to, FVIII activity in people treated with FVIII
 - Thrombin generation (TG) peak height was measured with two sets of TG assays, triggered by activated factor XI (FXIa) or tissue factor (TF)[3]
 - Endogenous FVIII activity was measured using a chromogenic assay with bovine factors (Siemens)
 - FVIII antigen levels were measured using an enzyme-linked immunosorbent assay (Affinity Biologicals).
- Genetic analyses: *F8* coding regions and exon/intron boundaries were sequenced using Sanger sequencing[4] and rearrangements were analyzed using inverse-shifting polymerase chain reaction.[5] Variants were mapped against population and disease genetics databases.[6–8]
- Safety analyses: Plasma levels of exploratory safety biomarkers (fibrinogen, prothrombin fragment 1.2 and D-dimer) were determined.[3]

Pharmacodynamic, genetic, and safety biomarker analyses were conducted in 72 participants

- At the data cut-off (30 October 2021), the median emicizumab follow-up period was 55.6 (interquartile range: 52.5–61.6) weeks.
- For a full list of participant baseline characteristics, please refer to Hermans *et al.*[9]

Pharmacodynamic biomarkers

Mean FVIII-like activity was sustained during exposure

- Mean FVIII-like activity increased during the loading phase, and was then sustained throughout emicizumab exposure.

Figure 1. Mean FVIII-like activity during emicizumab exposure

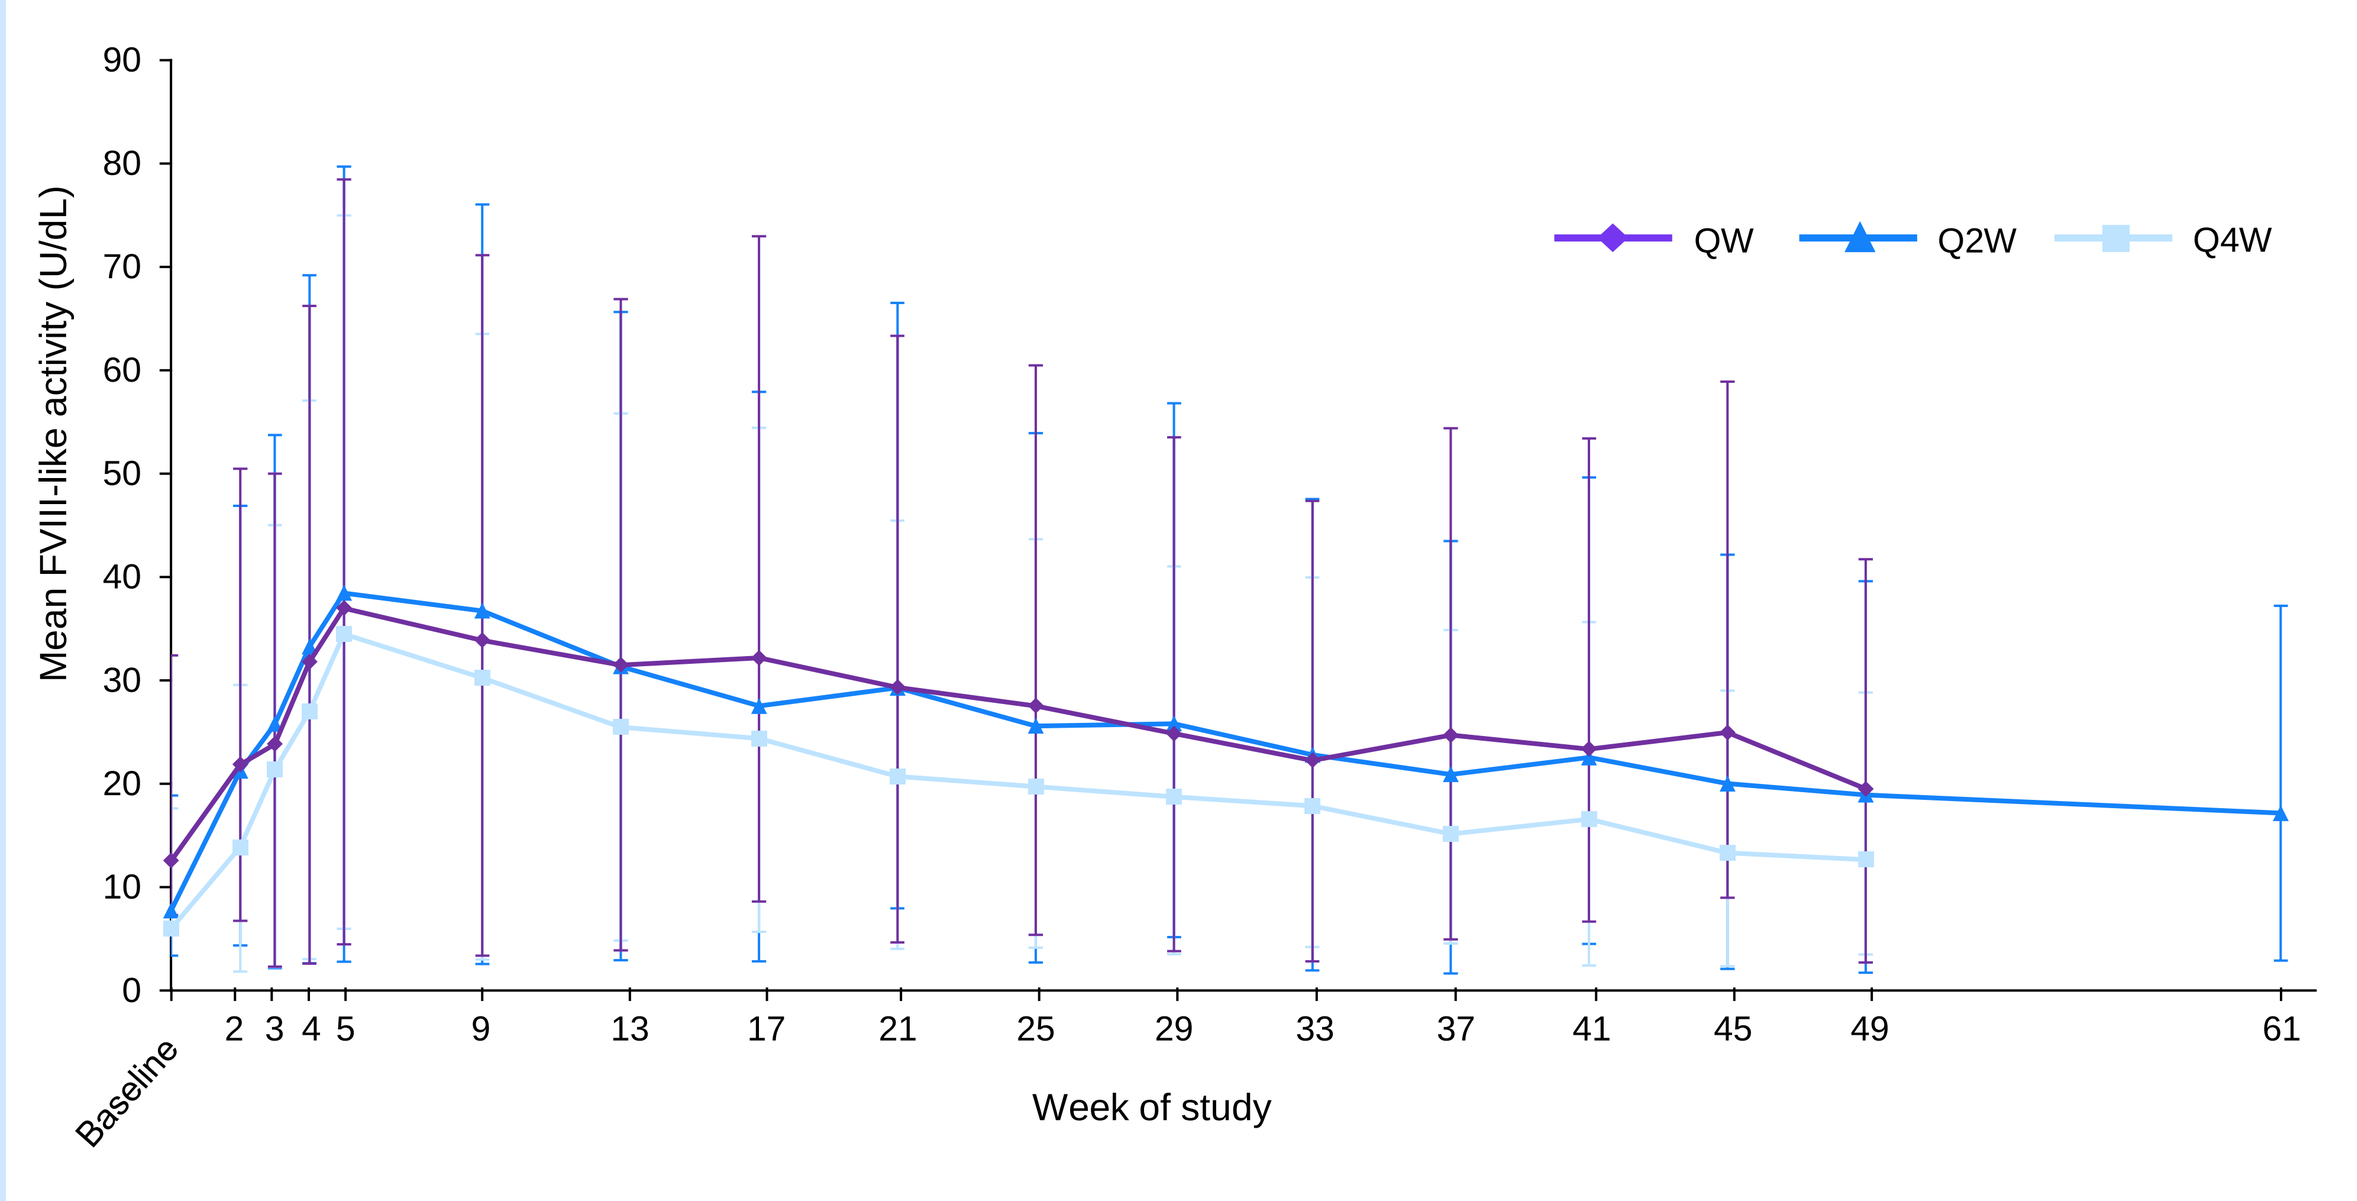
Mean FVIII-like activity (U/dL)
Week of study
Baseline 2 3 4 5 9 13 17 21 25 29 33 37 41 45 49 61
QW Q2W Q4W

Error bars represent 95% confidence interval. Normal range for FVIII activity: 50–150 U/dL.

aPTT was normalized after the first emicizumab dose

- Activated partial thromboplastin time (aPTT) remained stable during emicizumab exposure.

Figure 2. Mean aPTT levels during emicizumab exposure

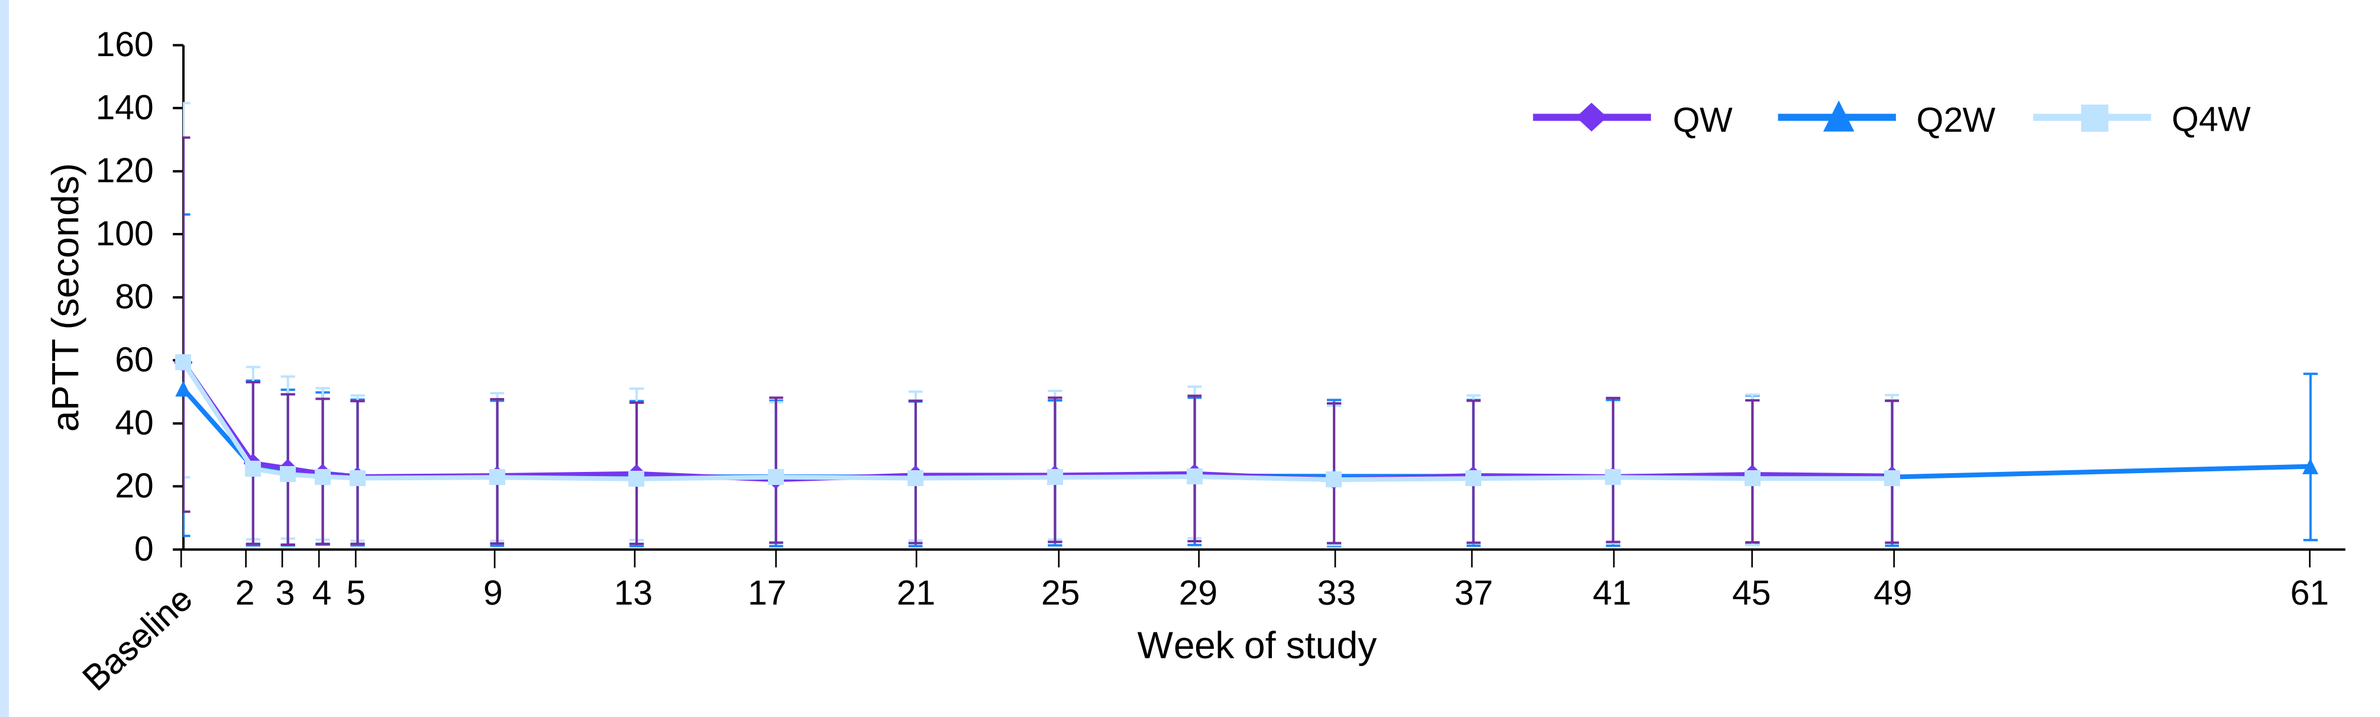
aPTT (seconds)
Week of study
QW Q2W Q4W

Error bars represent 95% confidence interval. Analysis conducted and reference ranges determined locally.

Mean TG peak height was maintained during exposure

- Mean TG peak height increased during the loading phase and was maintained during exposure, with levels below normal ranges.

Figure 3. Mean TG peak height during emicizumab exposure (A) FXIa trigger and (B) TF trigger

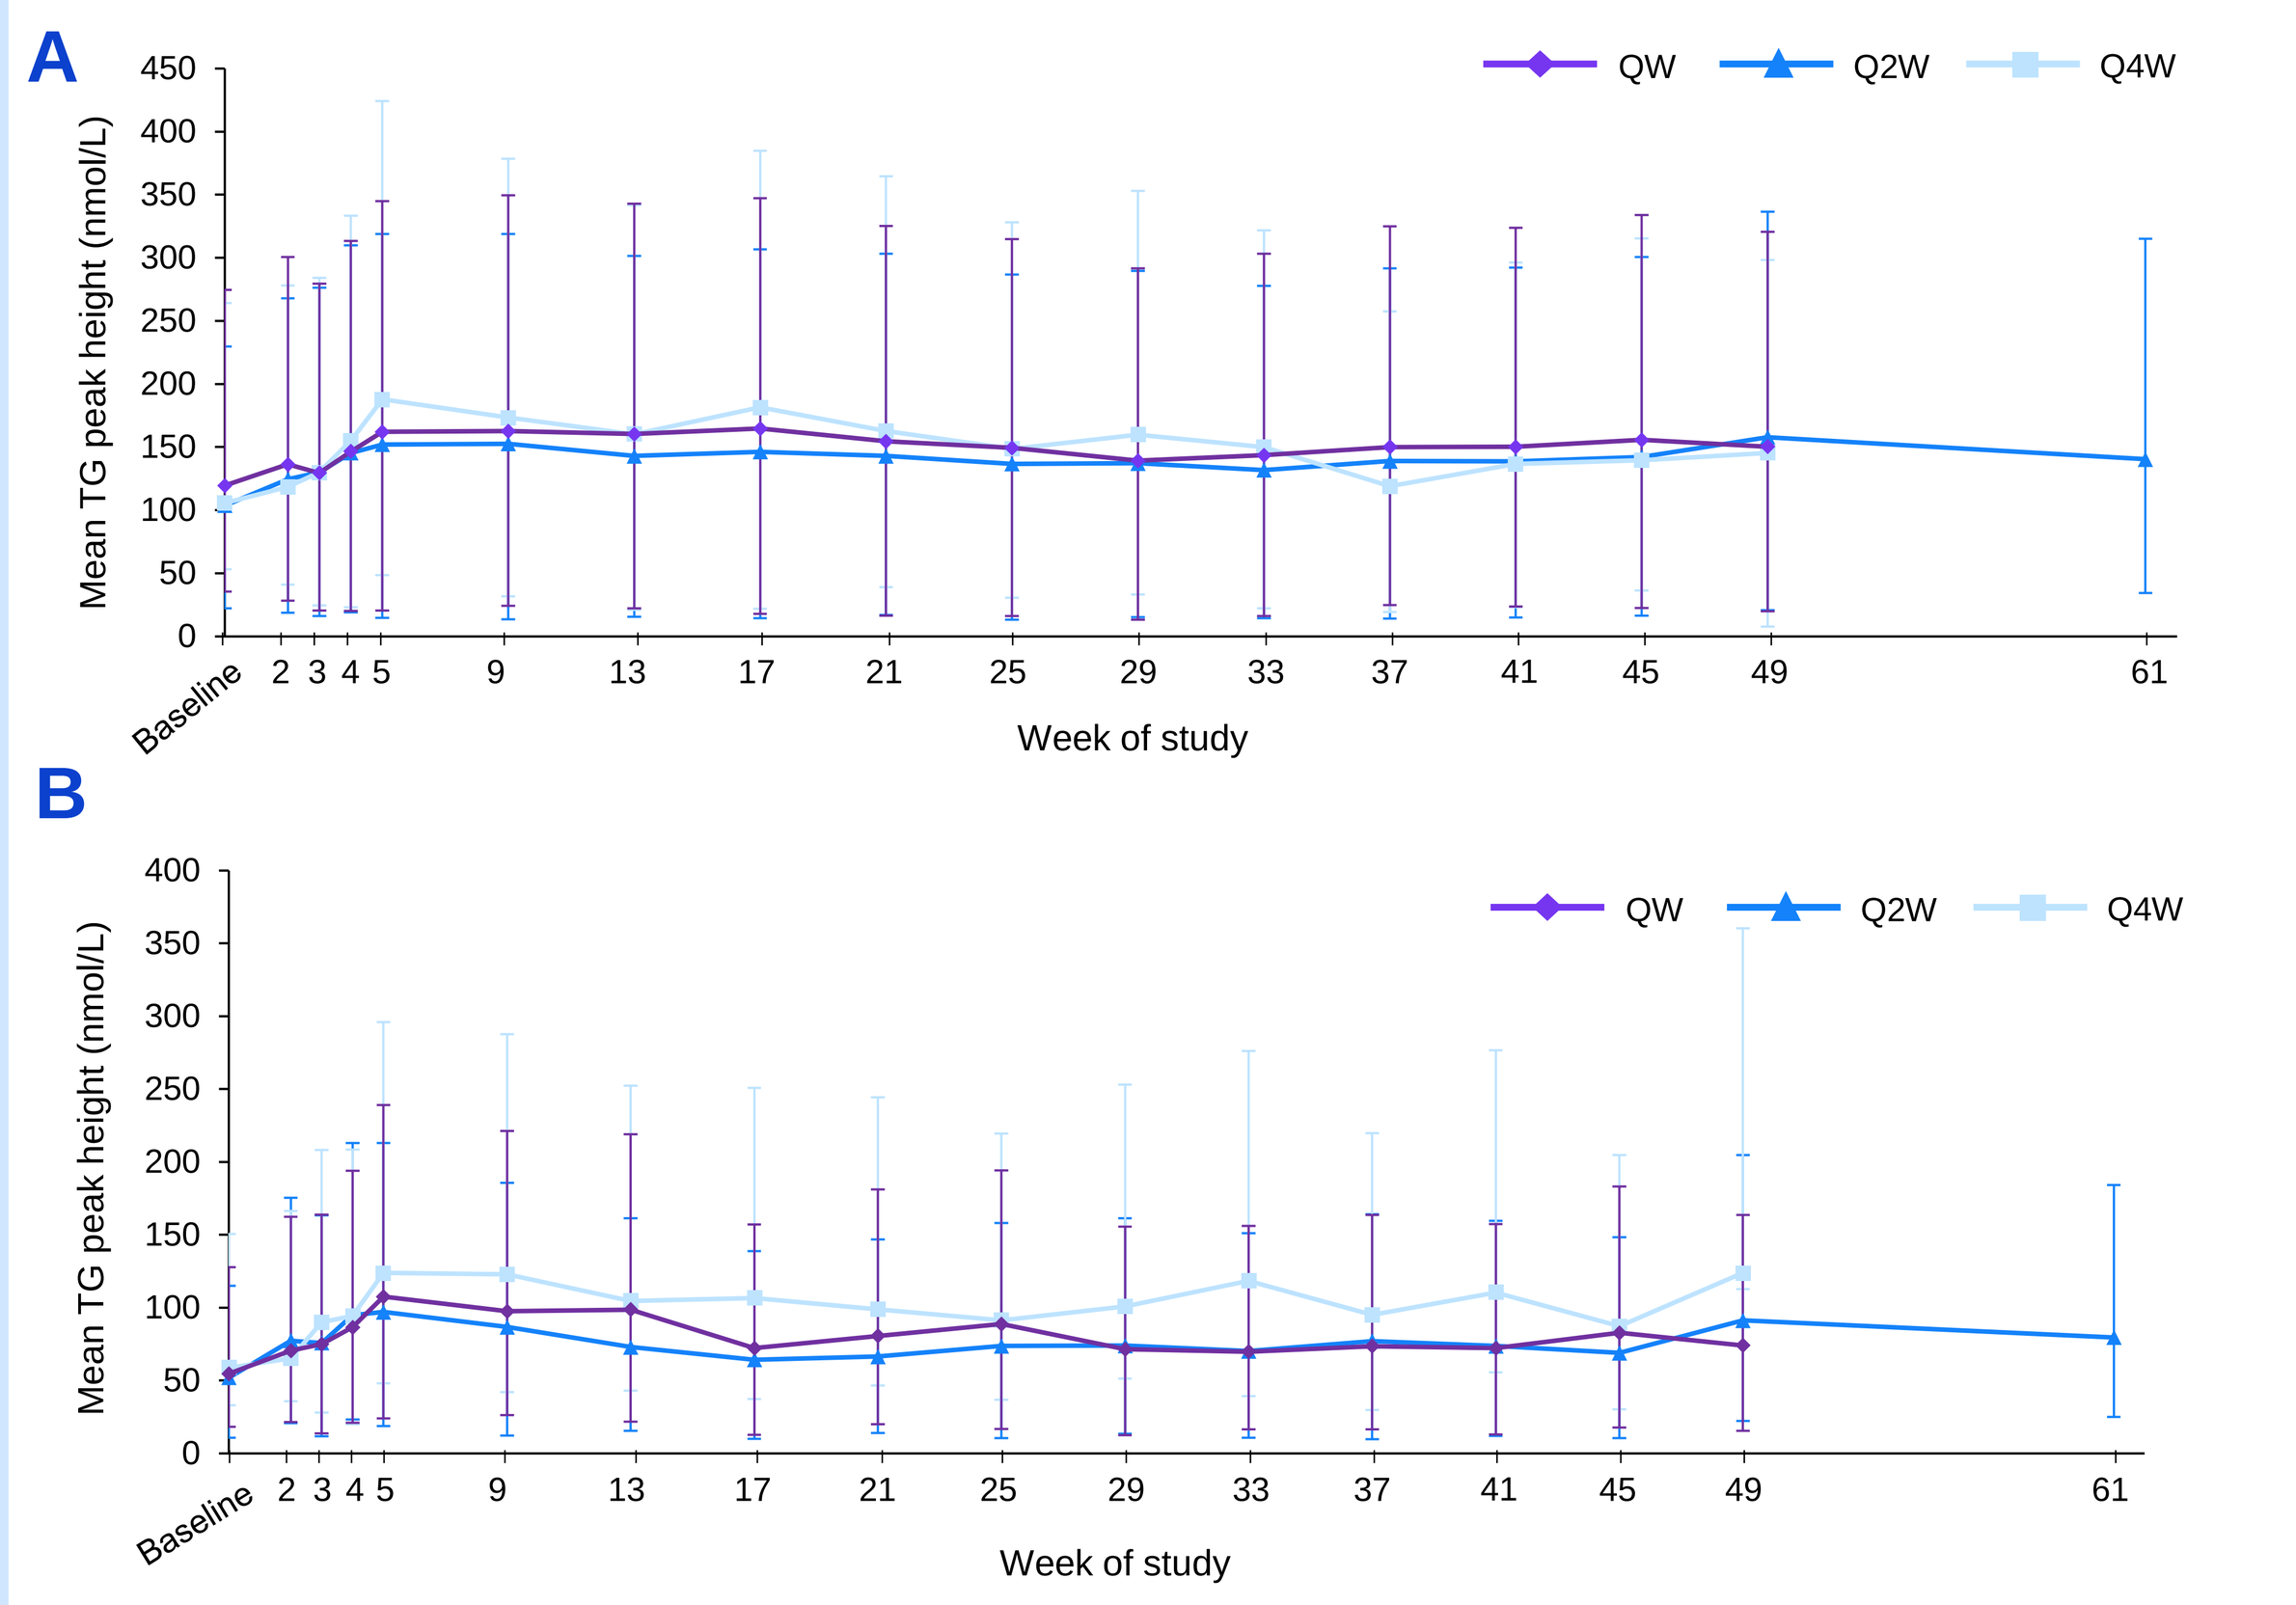
A
B
Mean TG peak height (nmol/L)
Week of study
QW Q2W Q4W

Error bars represent 95% confidence interval. TF trigger reference range:148–420 nmol/L. FXIa trigger reference range not determined.

Endogenous FVIII activity and FVIII antigen levels were unaffected by emicizumab

Figure 4. Individual (A) endogenous FVIII activity and (B) FVIII antigen levels during emicizumab exposure

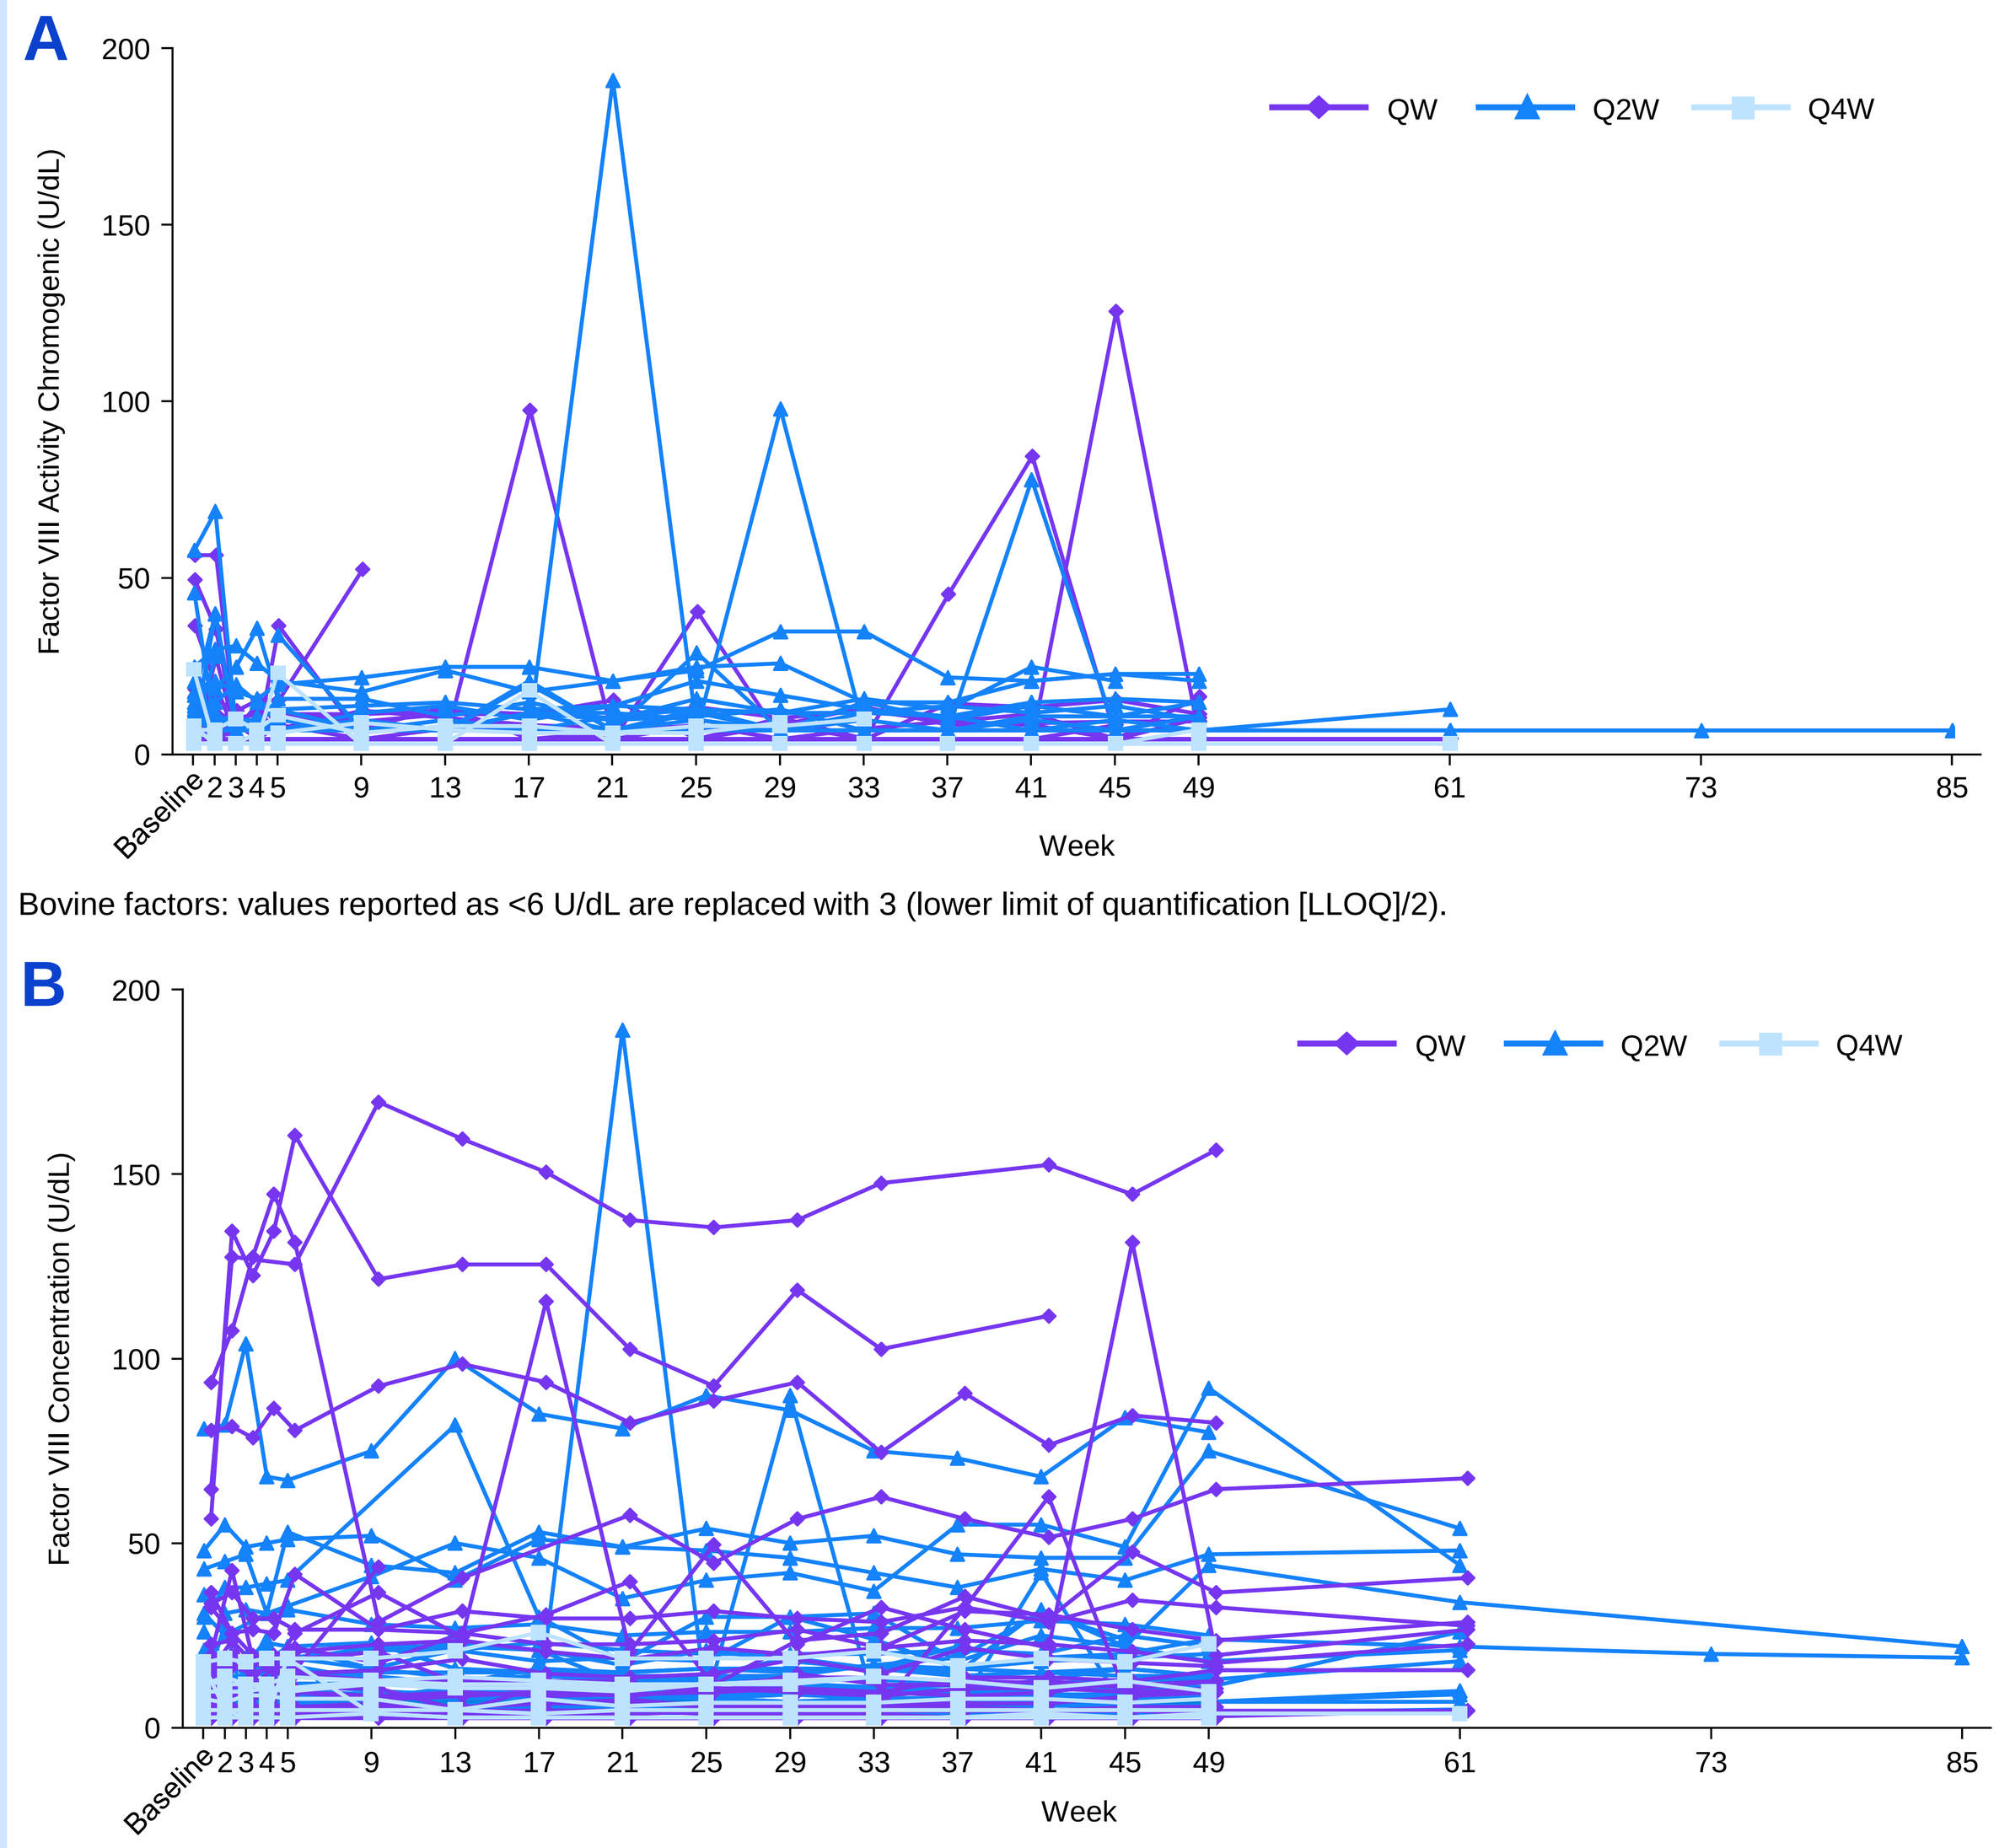
A
Factor VIII Activity Chromogenic (U/dL)
Week
QW Q2W Q4W
Bovine factors: values reported as <6 U/dL are replaced with 3 (lower limit of quantification [LLOQ]/2).
B
Factor VIII Concentration (U/dL)
Week
QW Q2W Q4W

Peaks within the graphs reflect FVIII use at baseline and sporadically to treat bleeds during the study.

Genetic biomarkers

Missense variants were the most common mutation in HAVEN 6 participants

- Overall, two rearrangements and 67 unique short variants were identified: 13 benign; 46 likely pathogenic/unclassified; and eight novel (i.e., variants not previously documented in genetic databases)
 - After excluding benign variants, 58 participants (81%) had one missense variant.

Figure 5. Variant type frequencies in HAVEN 6 participants

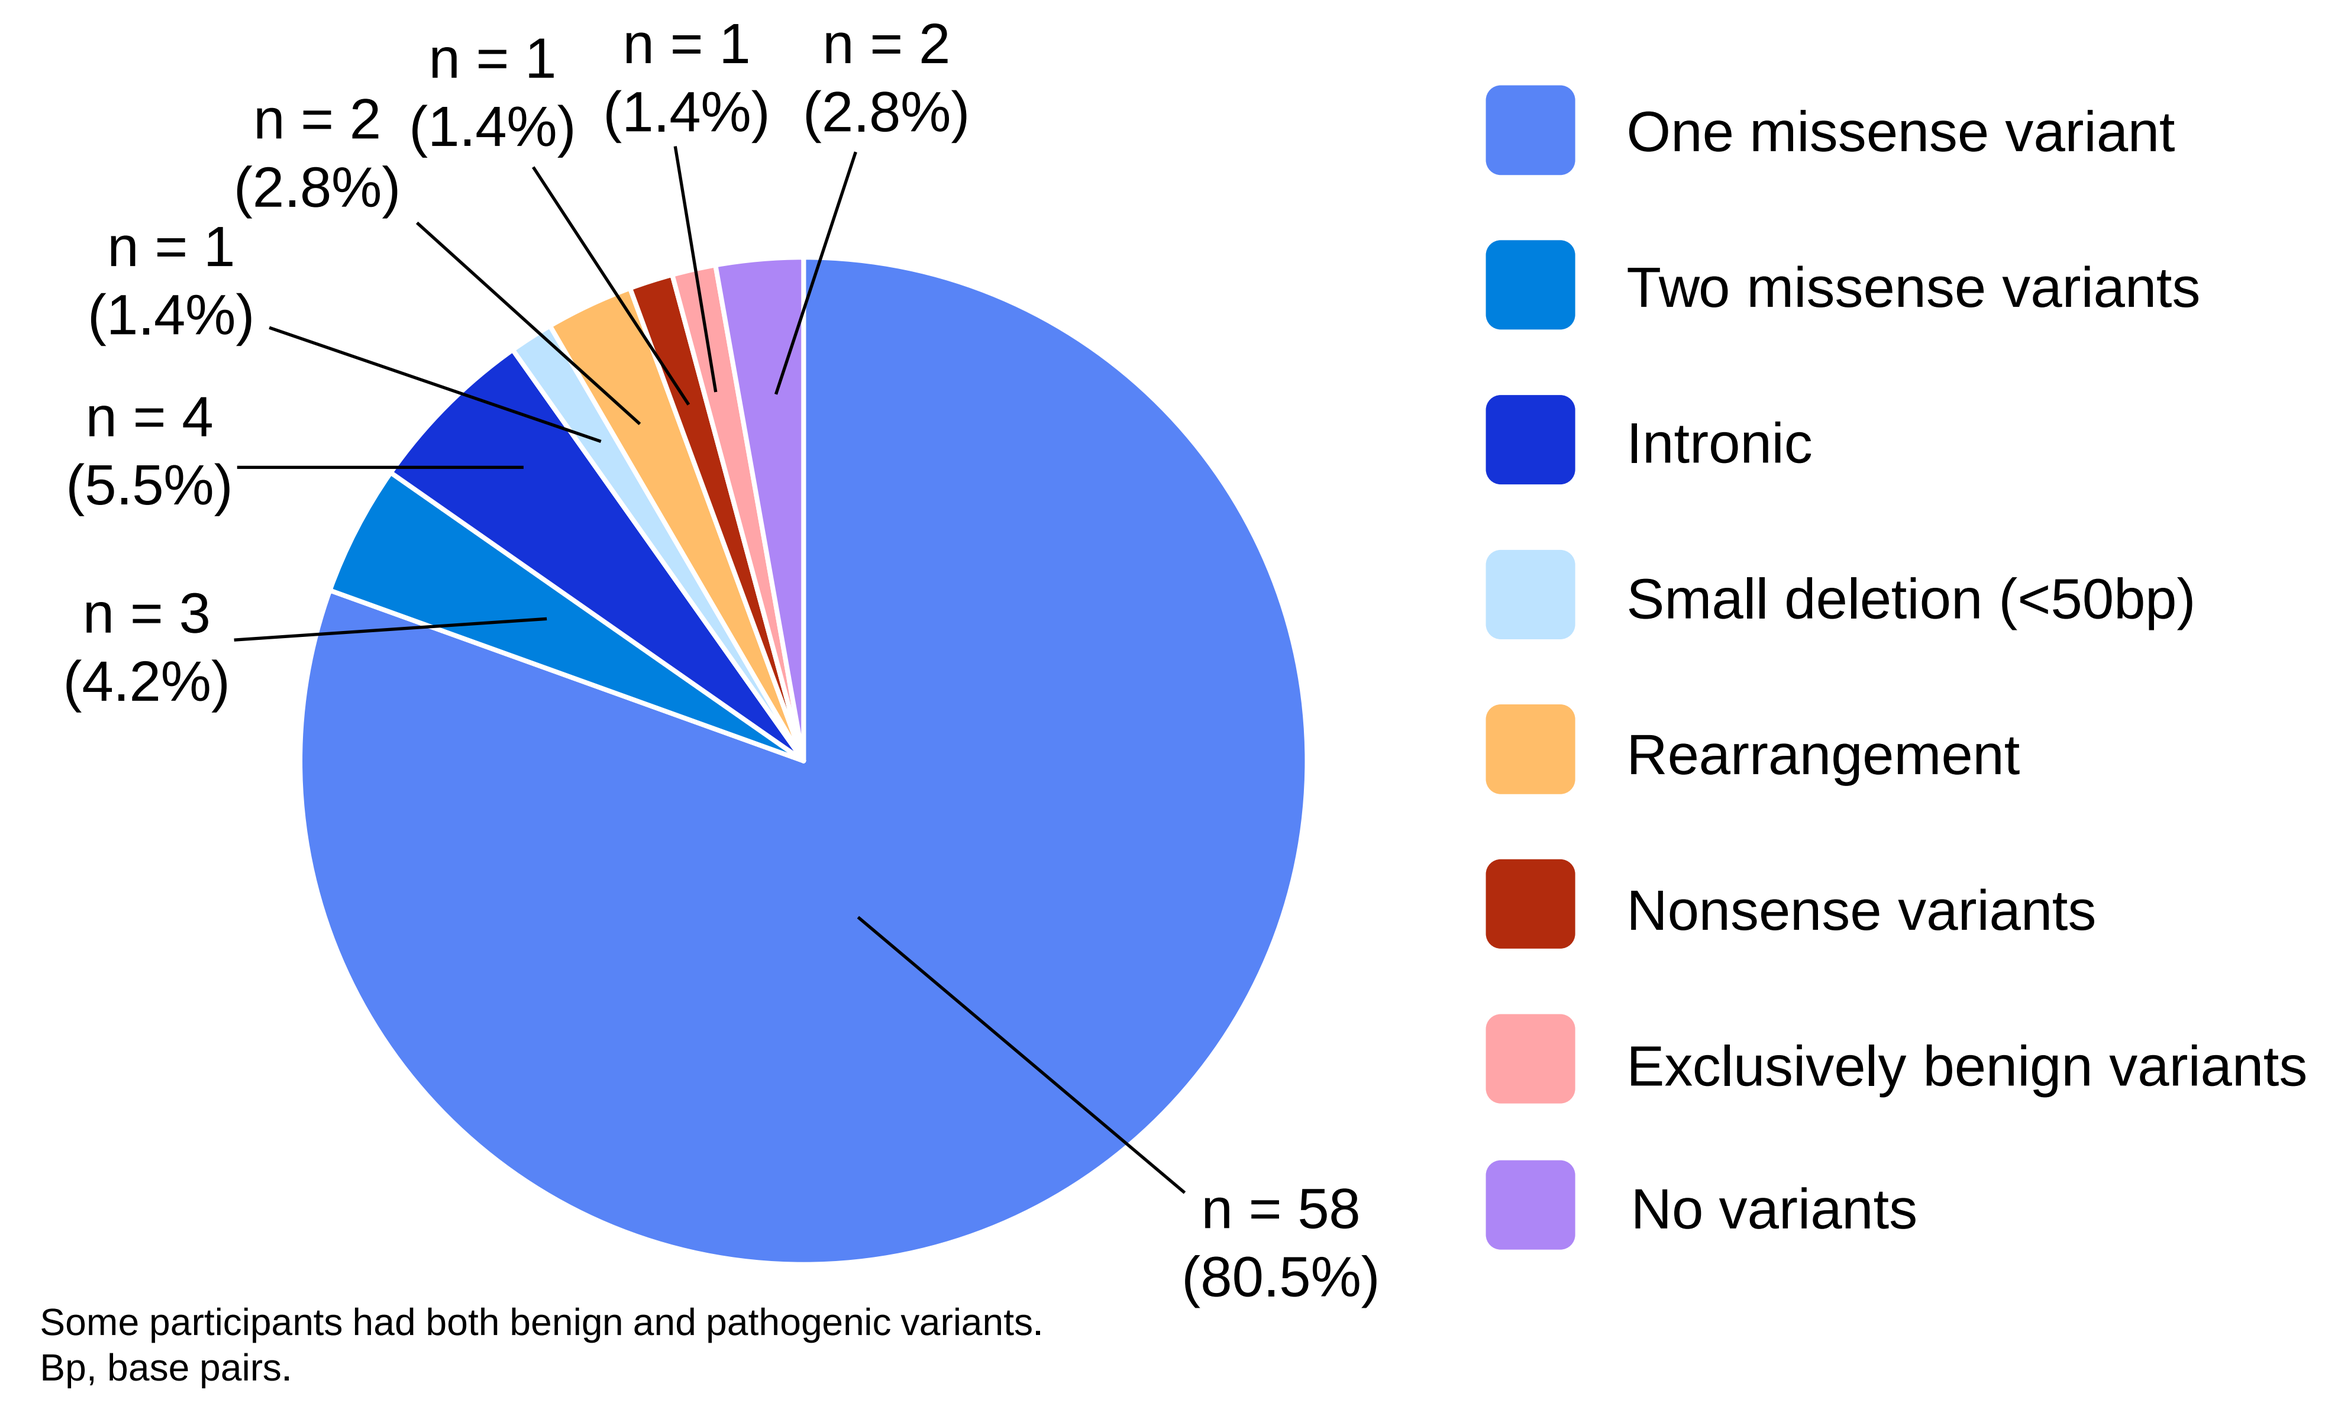
n = 58 (80.5%)
n = 3 (4.2%)
n = 4 (5.5%)
n = 1 (1.4%)
n = 2 (2.8%)
n = 1 (1.4%)
n = 1 (1.4%)
n = 2 (2.8%)
One missense variant
Two missense variants
Intronic
Small deletion (<50bp)
Rearrangement
Nonsense variants
Exclusively benign variants
No variants

Some participants had both benign and pathogenic variants.
Bp, base pairs.

Genetic variants were mapped against endogenous FVIII activity

- Both participants with a history of FVIII inhibitors had variants that have previously been reported in people with FVIII inhibitors.[6]

Figure 6. *F8* variants versus endogenous FVIII activity in all participants

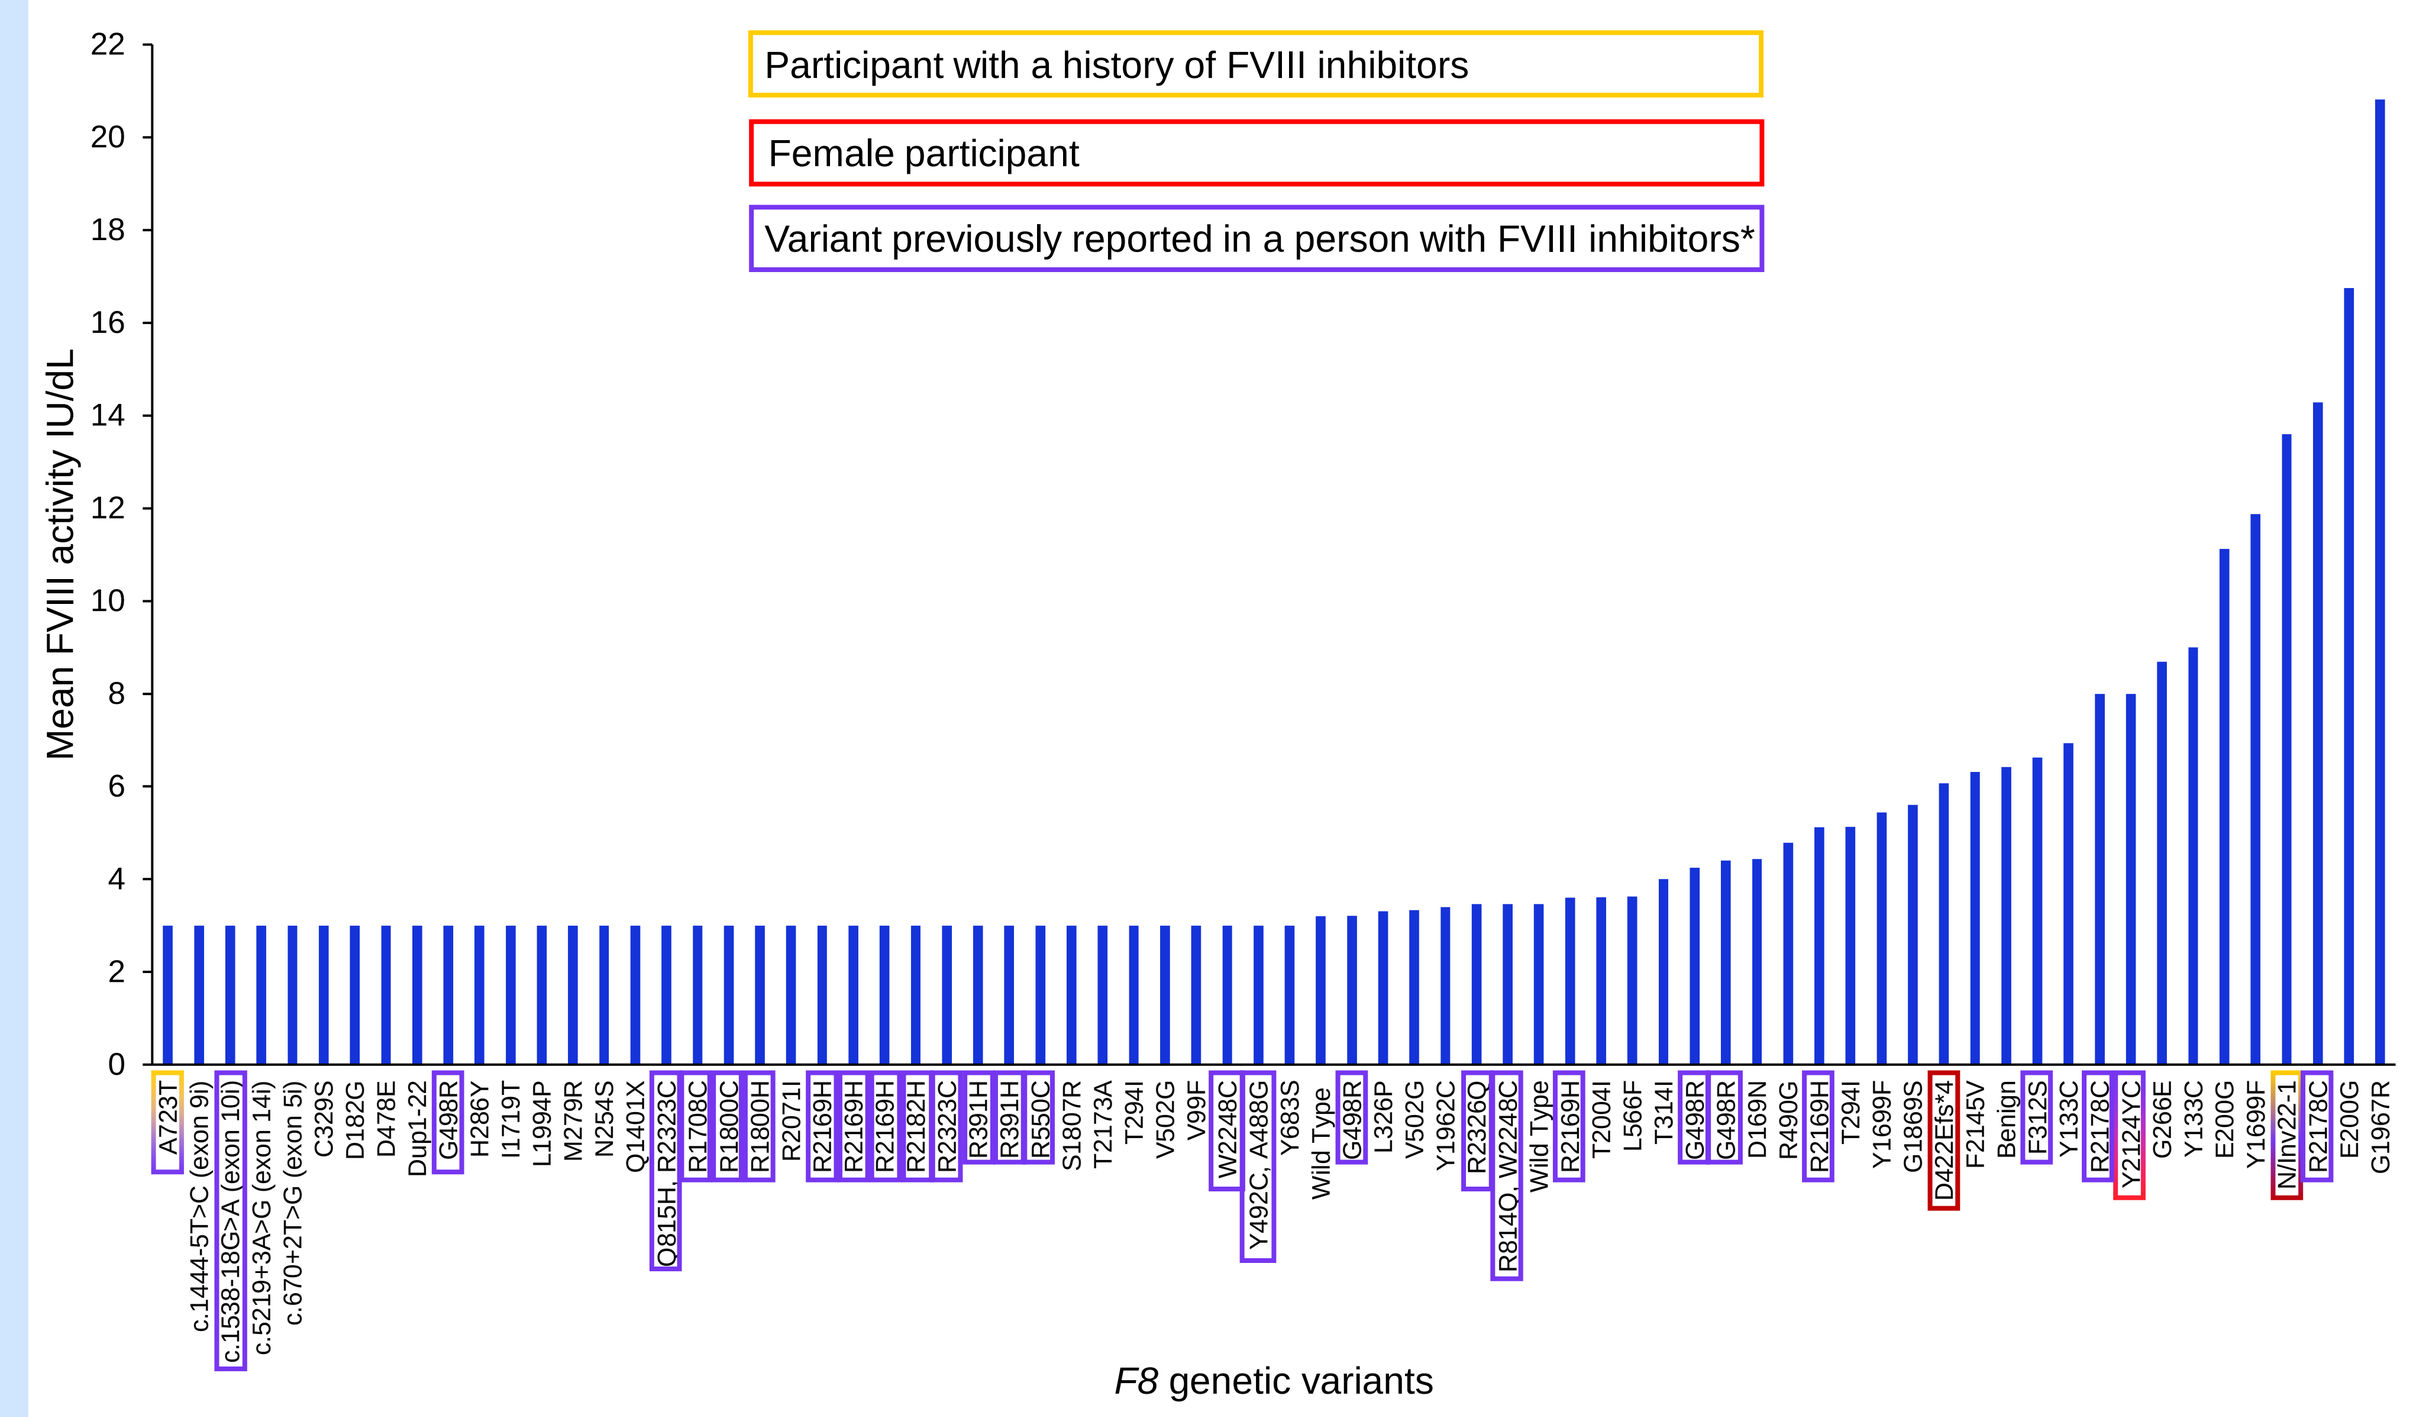
Participant with a history of FVIII inhibitors
Female participant
Variant previously reported in a person with FVIII inhibitors*
Mean FVIII activity IU/dL
F8 genetic variants
A723T, c.1444-5T>C (exon 9), c.1538-18G>A (exon 10), c.5219+3A>G (exon 14), c.670+2T>G (exon 5), C329S, D182G, D478E, Dup1-22, G498R, H286Y, I1719T, L1994P, M279R, N254S, Q1401X, Q815H, R2323C, R1708C, R1800C, R1800H, R2071I, R2169H, R2169H, R2169H, R2182H, R2323C, R391H, R391H, R550C, S1807R, T2173A, T294I, V502G, V99F, W2248C, Y492C, A488G, Y683S, Wild Type, G498R, L326P, V502G, Y1962C, R2326Q, R814Q, W2248C, Wild Type, R2169H, T2004I, L566F, T314I, G498R, G498R, D169N, R490G, R2169H, T294I, Y1699F, G1869S, D422Efs*4, F2145V, Benign, F312S, Y133C, R2178C, Y2124YC, G266E, Y133C, E200G, Y1699F, N/Inv22-1, R2178C, E200G, G1967R

*According to the European Association for Haemophilia and Allied Disorders (EAHAD) *F8* database.[6]
Mean FVIII activity across timepoints for each individual. Values <6 IU/dL (LLOQ of the assay) were set to three (LLOQ/2) for the calculation. Timepoints corresponding with FVIII treatment were excluded. The participant with only benign variants is indicated by 'benign'; this participant had three benign variants. Some participants had both pathogenic and benign variants; in these cases, only the suspected pathogenic variant(s) is included here. Two participants had no identifiable variants, and are indicated by 'wild type'. The Y2124YC heterozygous variant belongs to a female participant.

Safety biomarkers

Safety biomarkers were unaffected by emicizumab

- The safety biomarkers fibrinogen, prothrombin fragment 1.2 and D-dimer remained unchanged throughout emicizumab exposure.

Conclusions

- The identified *F8* mutations were consistent with mutations expected in people with moderate or mild HA.
- Emicizumab prophylaxis improved coagulation parameters (aPTT, FVIII-like activity, TG peak height) without exceeding normal ranges, impacting endogenous FVIII or affecting safety biomarkers.

Presented at the 2022 International Society on Thrombosis and Haemostasis (ISTH) Annual Meeting | July 9–13 2022

▼ This medicinal product is subject to additional monitoring. This will allow quick identification of new safety information. Healthcare professionals are asked to report any suspected adverse reactions. These should be reported to the Regulatory Authorities in your country according to your national requirements.

References

1. Kitazawa T, et al. *Thromb Haemost* 2017;117:1348–57.
2. Negrier C, et al. *Blood* 2021;138:343.
3. Schmitt C, et al. *Thromb Haemost* 2021;121:351–60
4. Vidal F, et al. *Thromb Haemost* 2001;85:580–3.
5. Rossetti LC, et al. *J Thromb Haemost* 2008;6:830–6.
6. *F8* database. Available at: https://f8-db.eahad.org/. [Accessed May 2022].
7. ClinVar database. Available at: https://www.clinicalgenome.org/data-sharing/clinvar/. [Accessed May 2022].
8. GnomAD database. Available at: https://gnomad.broadinstitute.org/. [Accessed May 2022].
9. Hermans C, et al. Presentation at ISTH 2022: OC 30.5.

Acknowledgments

The authors would like to thank the study participants and their families, as well as the study investigators, research coordinators and nurses, and Marilyn Barthoulot, who is the biomarker operations manager for the HAVEN 6 study. Medical writing support for the development of this poster, under the direction of the authors, was provided by Paige Phillips, MSc, of Ashfield MedComms, an Ashfield Health company, and funded by F. Hoffmann-La Roche Ltd.

Disclosures

AK is an employee of and stockholder in F. Hoffmann-La Roche Ltd.; CN has served as a consultant to BioMarin, Novo Nordisk, F. Hoffmann-La Roche Ltd. / Genentech, Inc., Sanofi, SOBI, and Shire/Takeda; has received research funding from Novo Nordisk, F. Hoffmann-La Roche Ltd. / Genentech, Inc., Sanofi, and SOBI; honoraria from Bayer, Novo Nordisk, F. Hoffmann-La Roche Ltd. / Genentech, Inc., Sanofi, SOBI, Spark, and Shire/Takeda; and has been a member of an entity's Board of Directors or advisory committees for Bayer, BioMarin, CSL Behring, Novo Nordisk, Pfizer, F. Hoffmann-La Roche Ltd. / Genentech, Inc., Sanofi, SOBI, Spark, Shire/Takeda, and UniQure; SWP has served as a consultant to Apcintex, ASC Therapeutics, Bayer, BioMarin, Catalyst Biosciences, CSL Behring, GenVentiv, HEMA Biologics, Freeline, Novo Nordisk, Pfizer, F. Hoffmann-La Roche Ltd. /Genentech, Inc., Sangamo Therapeutics, Sanofi, Takeda, Spark Therapeutics, UniQure; BMB is an employee and stockholder in F. Hoffmann-La Roche Ltd.; OC is an employee of F. Hoffmann-La Roche Ltd.; CS is an employee and stockholder in F. Hoffmann-La Roche Ltd., and is co-inventor of a patent related to an anti-FIXa/FX bispecific antibody; CH has received research funding from Bayer, Shire/Takeda, Pfizer, Novo Nordisk, CSL Behring, and SOBI; honoraria and speaker's bureau from Bayer, Shire/Takeda, Pfizer, Novo Nordisk, CSL Behring, Octapharma, SOBI, LFB, CAF-DCF, F. Hoffmann-La Roche Ltd., UniQure, and BioMarin; JO has received research funding from Bayer, Biotest, Chugai Pharmaceutical Co., Ltd., CSL Behring, Novo Nordisk, Octapharma, Pfizer, F. Hoffmann-La Roche Ltd., Swedish Orphan Biovitrum, and Takeda; consultancy, speakers bureau, honoraria, scientific advisory board, and travel expenses from Bayer, Biogen Idec, BioMarin, Biotest, Chugai Pharmaceutical Co., Ltd., CSL Behring, Freeline, Grifols, Novo Nordisk, Octapharma, Pfizer, F. Hoffmann-La Roche Ltd., Sanofi, Spark Therapeutics, Swedish Orphan Biovitrum, and Takeda; ML is an employee and holds stock in F. Hoffmann-La Roche Ltd.