P3999

Factor VIII Use in the Treatment of Breakthrough Bleeds in People with Moderate or Mild Hemophilia A without Factor VIII Inhibitors Receiving Emicizumab Prophylaxis: The Phase III HAVEN 6 Experience

Víctor Jiménez-Yuste,[1]* Elise Lim,[2] Ketna Mistry,[2] Giuliana Ventriglia,[3] Amy Shapiro,[4]

*presenting author

Summary

Data from the Phase III HAVEN 6 trial show a favorable safety profile and efficacy of emicizumab in people with moderate or mild hemophilia A (HA) without factor (F)VIII inhibitors

This sub-analysis characterizes the dose and frequency of FVIII and tranexamic acid (TxA) used for the treatment of breakthrough bleeds throughout the duration of the HAVEN 6 study

These data may be used to inform breakthrough bleed treatment in people with moderate or mild HA receiving emicizumab

Reported bleeds were mainly traumatic and were resolved after 1–2 doses of FVIII. No safety concerns were observed with concomitant use of emicizumab and TxA

Affiliations: [1]Hospital Universitario La Paz-IdiPaz, Universidad Autónoma, Madrid, Spain; [2]Genentech, Inc., South San Francisco, CA, USA; [3]F. Hoffmann-La Roche Ltd, Basel, Switzerland; [4]Indiana Hemophilia & Thrombosis Center, Indianapolis, IN, USA.

Background

- Emicizumab, a bispecific monoclonal antibody that bridges activated factor (F)IX and FX, substitutes for the function of deficient FVIII in people with hemophilia A (HA); data from the Phase III HAVEN 6 trial (NCT04158648) show a favorable safety profile and efficacy of emicizumab in people with moderate or mild HA without FVIII inhibitors.[1]
- This analysis characterizes the dose and frequency of FVIII, and tranexamic acid (TxA) used for the treatment of breakthrough bleeds in people with moderate or mild HA treated with emicizumab during the HAVEN 6 trial.

HAVEN 6 is a multicenter, open-label, single-arm, Phase III study designed to assess safety and efficacy of emicizumab in people with moderate or mild HA

- Eligible participants included people of all ages with moderate or mild HA without inhibitors, who warranted prophylaxis as assessed by the investigator; the full study design has been published.[1]
- The lowest FVIII dose required to achieve hemostasis was recommended for breakthrough bleeds.
- Self-reported bleeds were defined as per the International Society on Thrombosis and Haemostasis;[2] treated bleeds excluded surgical bleeds.
- TxA use in the 24 hours before a surgical procedure was excluded from this analysis.
- No formal statistical tests were performed.

Figure 1. Study design.

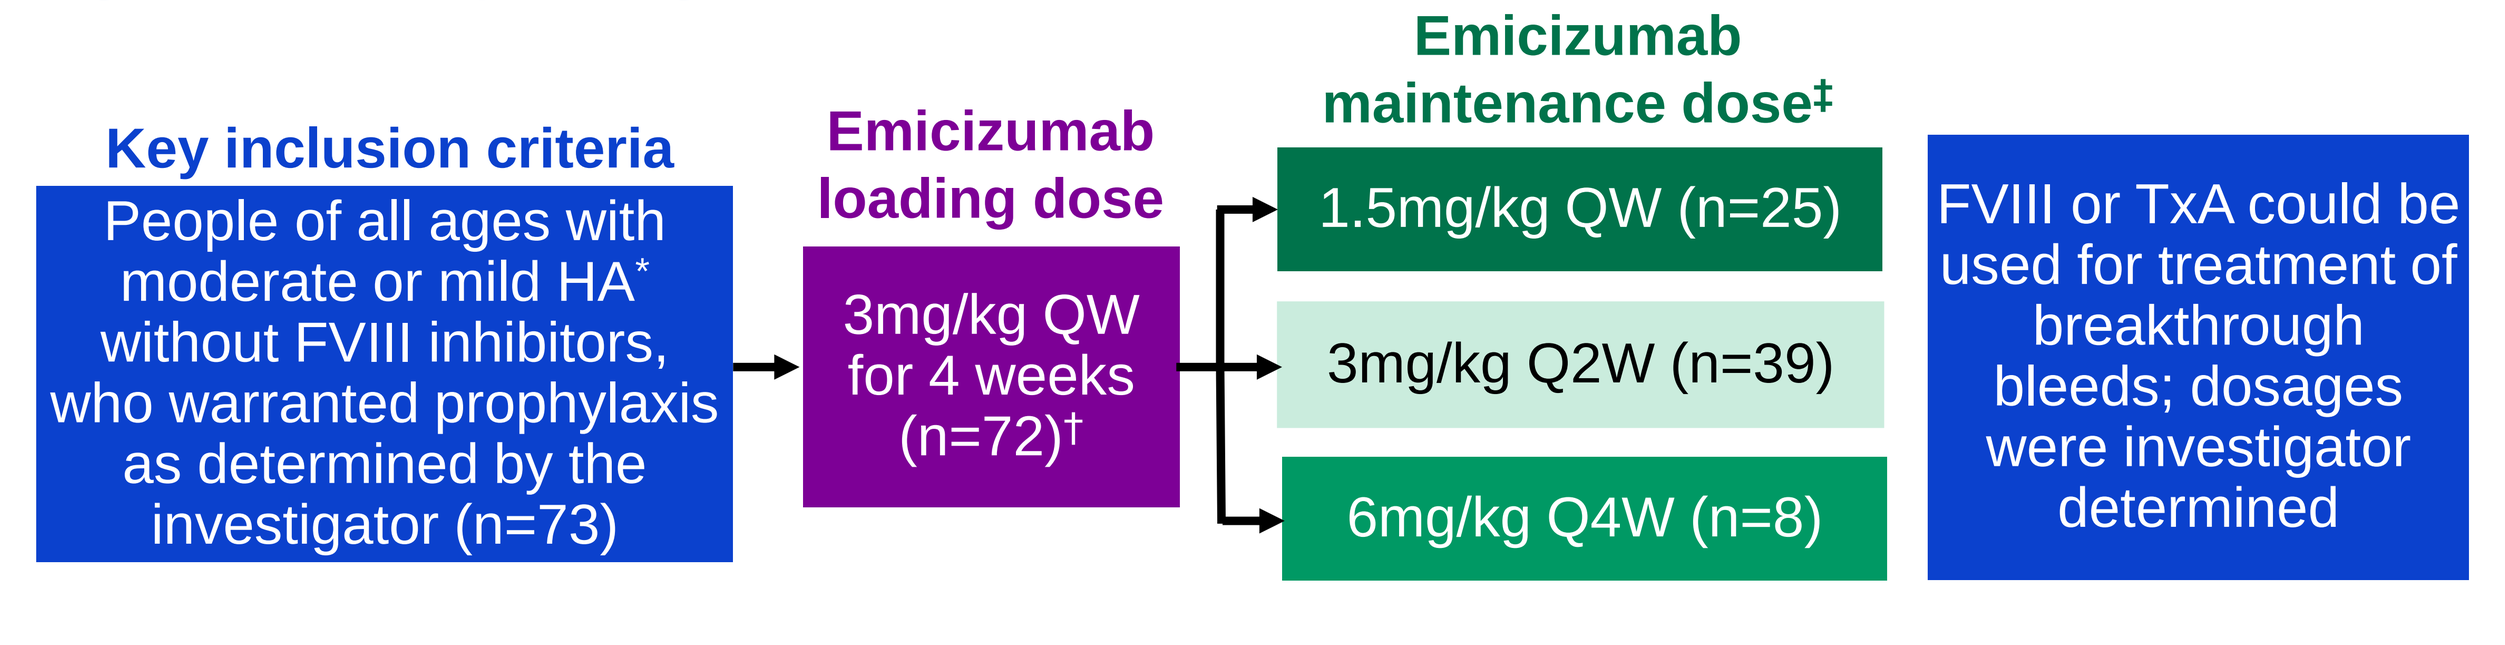

Key inclusion criteria
People of all ages with moderate or mild HA* without FVIII inhibitors, who warranted prophylaxis as determined by the investigator (n=73)
Emicizumab loading dose
3mg/kg QW for 4 weeks (n=72)†
Emicizumab maintenance dose‡
1.5mg/kg QW (n=25)
3mg/kg Q2W (n=39)
6mg/kg Q4W (n=8)
FVIII or TxA could be used for treatment of breakthrough bleeds; dosages were investigator determined

*Mild HA (FVIII level >5%–<40%), moderate HA (FVIII level ≥1%–≤5%); †One participant with moderate HA was enrolled but withdrew prior to treatment; ‡Participant choice. QW, every week; Q2W, every two weeks; Q4W, every four weeks.

A total of 72 participants received emicizumab by data cut-off

- At the data cut-off (October 30, 2021), the median (range) follow-up was 55.6 (8.7–89.9) weeks; baseline characteristics are presented in **Table 1.**

Table 1. Baseline characteristics.

	Participants in the HAVEN 6 study		
	Total (N=72)	Mild HA (n=21)	Moderate HA (n=51)
Age, years			
Mean (SD)	26.0 (17.1)	33.5 (19.0)	22.9 (15.5)
Median (min, max)	23.5 (2.0, 71.0)	29.0 (6.0, 71.0)	19.0 (2.0, 56.0)
Sex, n (%)			
Male	69 (95.8)	19 (90.5)	50 (98.0)
Female	3 (4.2)	2 (9.5)	1 (2.0)
Treatment regimen prior to study, n (%)			
Prophylactic	37 (51.4)	9 (42.9)	28 (54.9)
Episodic	35 (48.5)	12 (57.1)	23 (45.1)
History of FVIII inhibitor, n (%)			
Yes*	2 (2.8)	1 (4.8)	1 (2.0)
No	70 (97.2)	20 (95.2)	50 (98.0)
Number of bleeds in 24 weeks prior to study			
Mean (SD)	4.7 (13.2)	9.3 (23.7)	2.7 (3.1)
Median (min, max)	2.0 (0.0, 96.0)	1.0 (0.0, 96.0)	2.0 (0.0, 14.0)
0–8 bleeds	67 (93.1)	18 (85.7)	49 (96.1)
≥9 bleeds	5 (6.9)	3 (14.3)	2 (3.9)
Estimated ABR prior to study†			
Mean (SD)	10.1 (28.7)	20.2 (51.5)	6.0 (6.8)
Median (Q1, Q3)	4.3 (0.0, 9.8)	2.2 (0.0, 10.9)	4.3 (0.0, 8.7)
Target joints at study entry			
No target joints, n (%)	48 (66.7)	15 (71.4)	33 (64.7)
≥1 target joint, n (%)	24 (33.3)	6 (28.6)	18 (35.3)
Reason for warranting prophylaxis, n (%)‡			
History of frequent bleeding	41 (56.9)	12 (57.1)	29 (56.9)
History of frequent joint bleeding	32 (44.4)	7 (33.3)	25 (49.0)
History of severe bleeding	15 (20.8)	5 (23.8)	10 (19.6)
Prevention of traumatic bleeds	9 (12.5)	4 (19.0)	5 (9.8)
Other	5 (6.9)	1 (4.8)	4 (7.8)

*Participants had to have a negative test for inhibitors (<0.6BU/mL) within 8 weeks before enrollment and no documented inhibitors (<0.6BU/mL), FVIII half-life <6 hours, or FVIII recovery <66% in the last 5 years. The reported highest historical inhibitor titer for the two participants with a history of inhibitors was approximately 1BU/mL. †Estimated ABR prior to study entry was derived from the participants' number of bleeds in the 24 weeks prior to the study (collected retrospectively), annualized by dividing by 168 days and multiplied by 365.25 days. ‡More than one investigator-assessed reason could be selected. ABR, annualized bleed rate; Q, quartile; SD, standard deviation.

The majority of treated bleeds were defined as traumatic

- During the HAVEN 6 study, 48 (66.7%) participants did not experience a treated bleed.
- Of the 71 treated bleeds, 57 (80.3%) were in those with moderate HA and 14 (19.7%) were in those with mild HA.
- A total of 56 (78.9%) of the 71 treated bleeds were traumatic and 15 (21.1%) were spontaneous.
- Sixteen (31.4%) of the 51 participants with moderate HA and 8 (38.1%) of the 21 participants with mild HA received on-demand FVIII to treat a breakthrough bleed; all participants who experienced a bleed remained on emicizumab, regardless of additional FVIII usage.
- Four participants experienced ≥6 treated bleeds during the study; a total of 28 (90.3%) of the 31 bleeds experienced by these participants were traumatic
 - These participants all had moderate HA and were receiving either prophylactic treatment (n=1) or episodic treatment (n=3) prior to the study
 - Three spontaneous bleeds were experienced by two participants and were located in the right knee and left forearm for one participant and in the left chest (in relation to a thorax contusion) for the other participant.

The majority of breakthrough bleeds were treated with one or two infusions of FVIII

- Of the 71 breakthrough bleeds, 54 (76.1%) needed 1–2 FVIII infusions (**Figure 2**).
- Overall, the median (range) number of FVIII infusions per breakthrough bleed was 2 (1–47) and 1 (1–6) for participants with moderate and mild HA, respectively.

Figure 2. Number of FVIII infusions per treated bleed in the overall population (N=72).

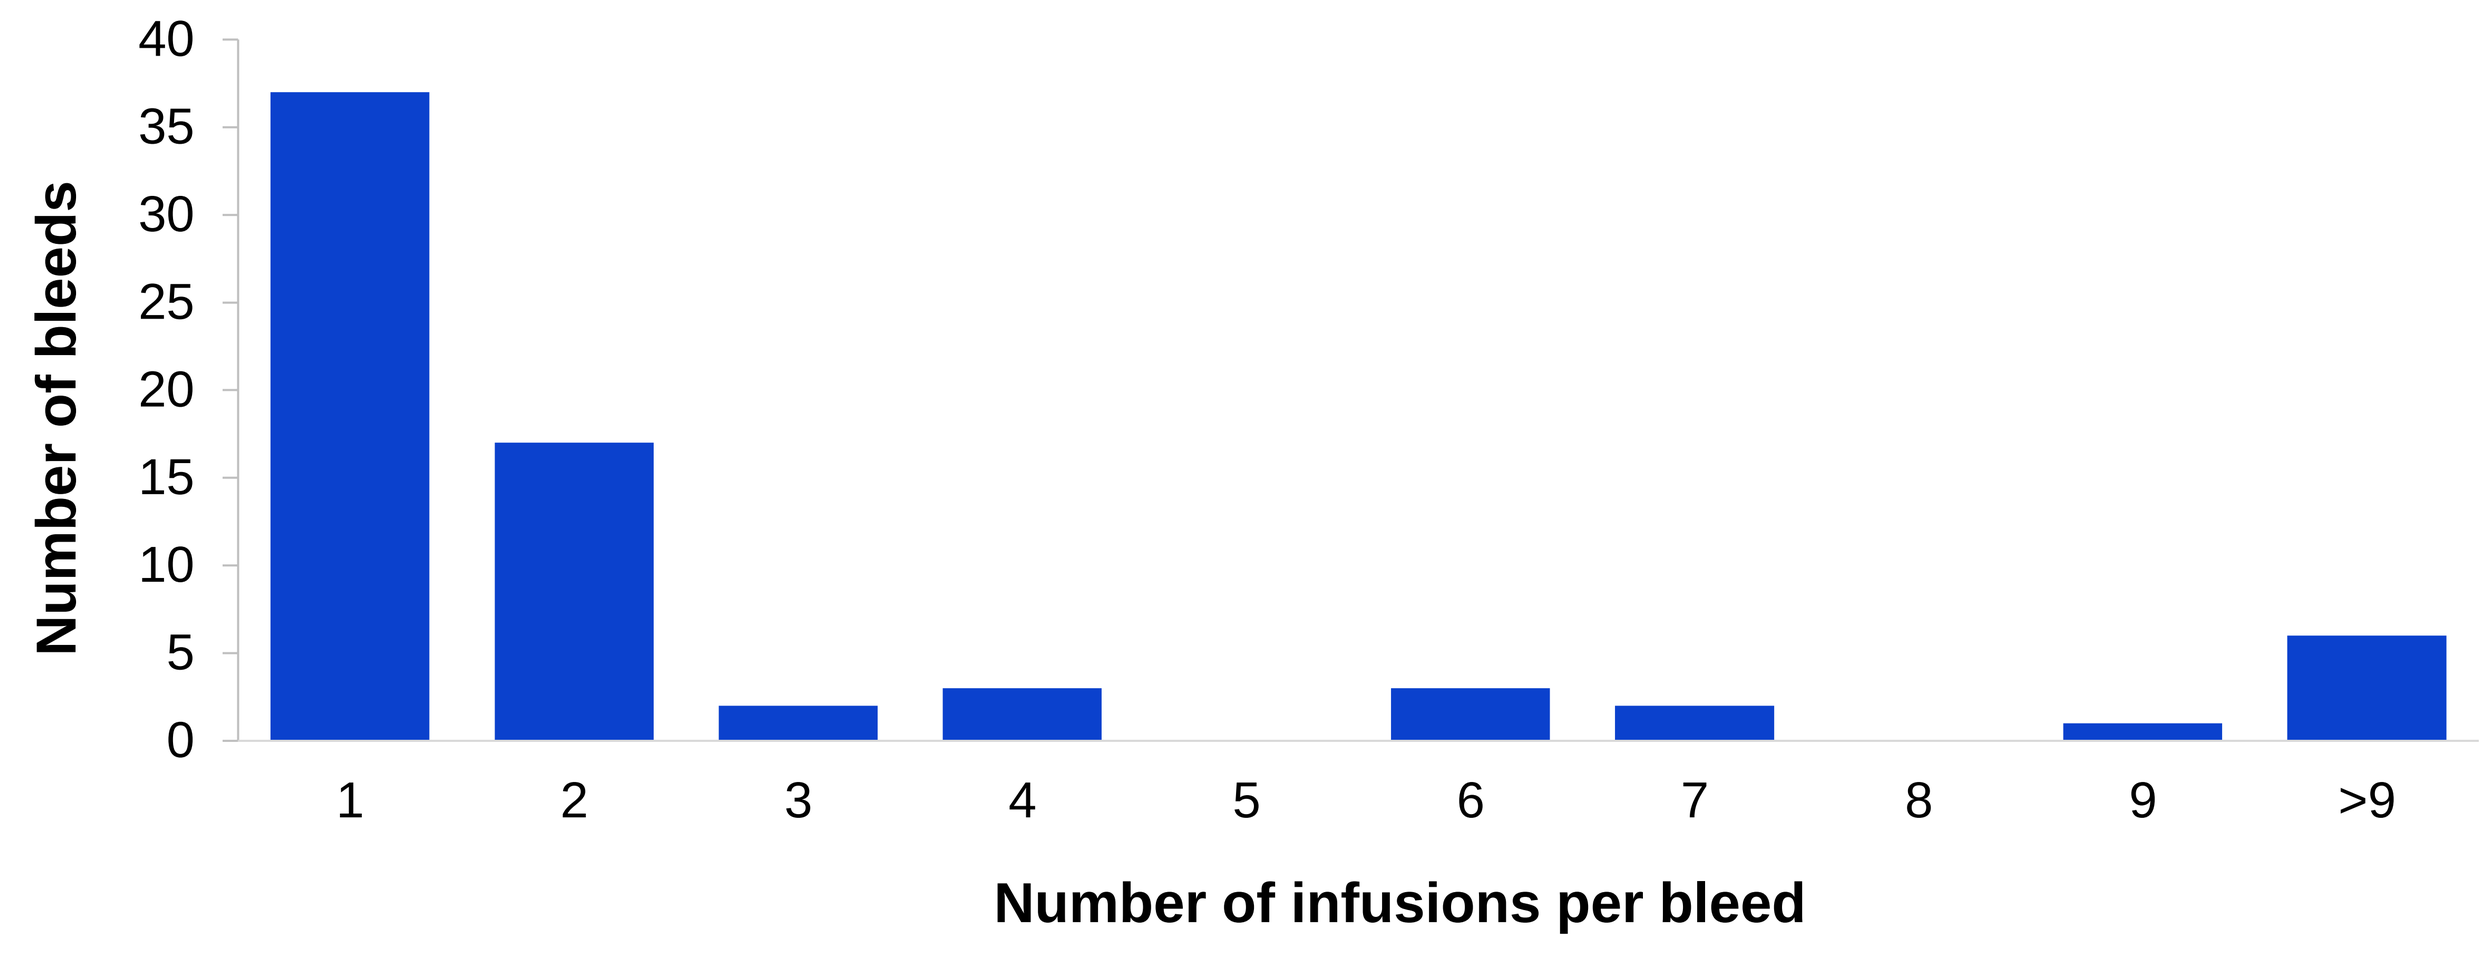

Number of bleeds
40
35
30
25
20
15
10
5
0
1
2
3
4
5
6
7
8
9
>9
Number of infusions per bleed

- The overall median (range) cumulative FVIII dose per bleed was 67.1 (31.5–170.0) IU/kg for those with moderate HA and 47.0 (40.1–50.5) IU/kg for those with mild HA (**Figure 3**)
 - Two participants (both aged 13 with moderate HA) required more than nine FVIII infusions to treat breakthrough bleeds, and are not represented in Figure 3
 - One participant had 19 infusions for a traumatic joint bleed on the finger/thumb.
 - The other participant experienced two traumatic bleeds in the thigh/knee, (treated with 47 infusions); the same participant experienced a spontaneous bleed in relation to a thorax contusion (treated with 19 infusions), a traumatic joint bleed on the left ankle treated with 18 infusions, and a traumatic joint bleed in the finger/thumb treated with 13 infusions.

Figure 3. Number of infusions and cumulative dose of FVIII per bleed in participants who received ≤9 infusions per bleed.

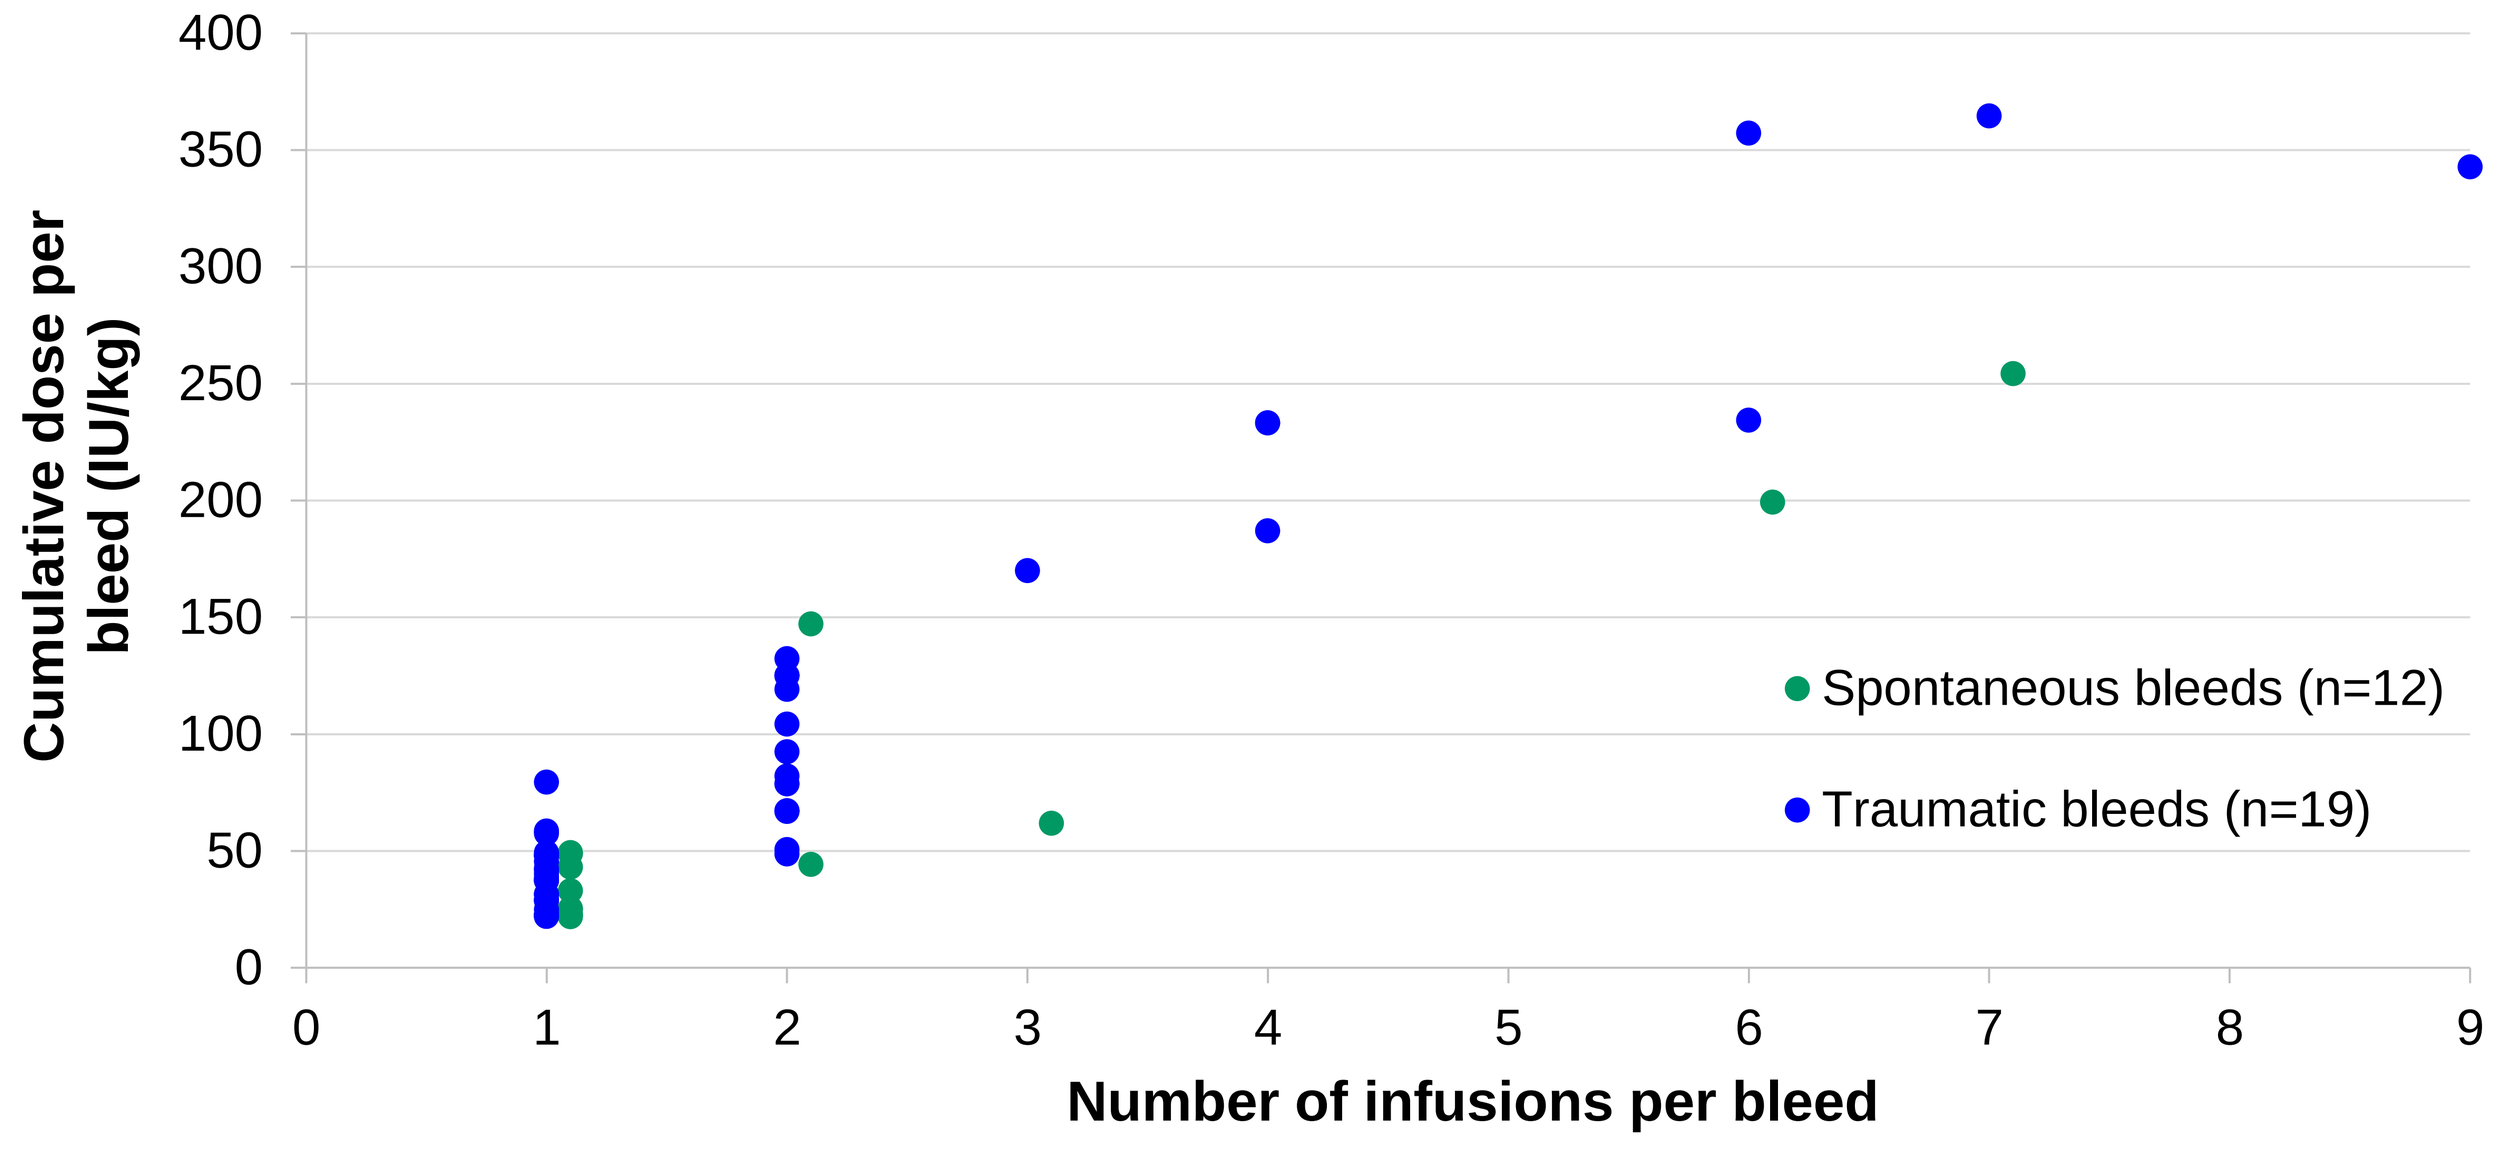

Cumulative dose per bleed (IU/kg)
400
350
300
250
200
150
100
50
0
0
1
2
3
4
5
6
7
8
9
Number of infusions per bleed
Spontaneous bleeds (n=12)
Traumatic bleeds (n=19)

A total of 12 participants experienced 14 spontaneous bleeds, and 19 participants experienced 51 traumatic bleeds.

The median number of annualized FVIII infusions per participant was 0 (0–120)

- The median (range) annualized number of infusions per participant was 0 (0–120) for those with moderate HA and 0 (0–18) for those with mild HA (**Table 2**).

Table 2. Annualized number of FVIII infusions per treated bleed per participant in the overall population (N=72).

	All bleeds	Spontaneous bleeds	Traumatic bleeds	Joint bleeds
Total population (N=72)				
Median (range)	0 (0–120.0)	0 (0–18.3)	0 (0–105.5)	0 (0–23.7)
Mean (SD)	3.5 (14.7)	0.8 (2.9)	2.7 (12.9)	0.8 (3.6)
Moderate HA (n=51)				
Median (range)	0 (0–120.0)	0 (0–14.5)	0 (0–105.5)	0 (0–23.7)
Mean (SD)	4.1 (17.2)	0.6 (2.2)	3.6 (15.3)	0.9 (4.05)
Mild HA (n=21)				
Median (range)	0 (0–18.3)	0 (0–18.3)	0 (0–5.4)	0 (0–10.7)
Mean (SD)	2 (4.7)	1.4 (4.2)	0.6 (1.3)	0.6 (2.3)

- For those with moderate HA, the median (range) number of FVIII infusions per treated spontaneous, traumatic and joint bleed was 1 (1–19), 2 (1–47) and 1 (1–19), respectively (**Figure 4**).
- For those with mild HA, the median (range) number of FVIII infusions per treated spontaneous, traumatic and joint bleed was 1 (1–6), 1 (1–2) and 2 (1–6), respectively (**Figure 4**).

Figure 4. Number of infusions required for the treatment of each type of treated bleed in participants who received ≤9 infusions per bleed.

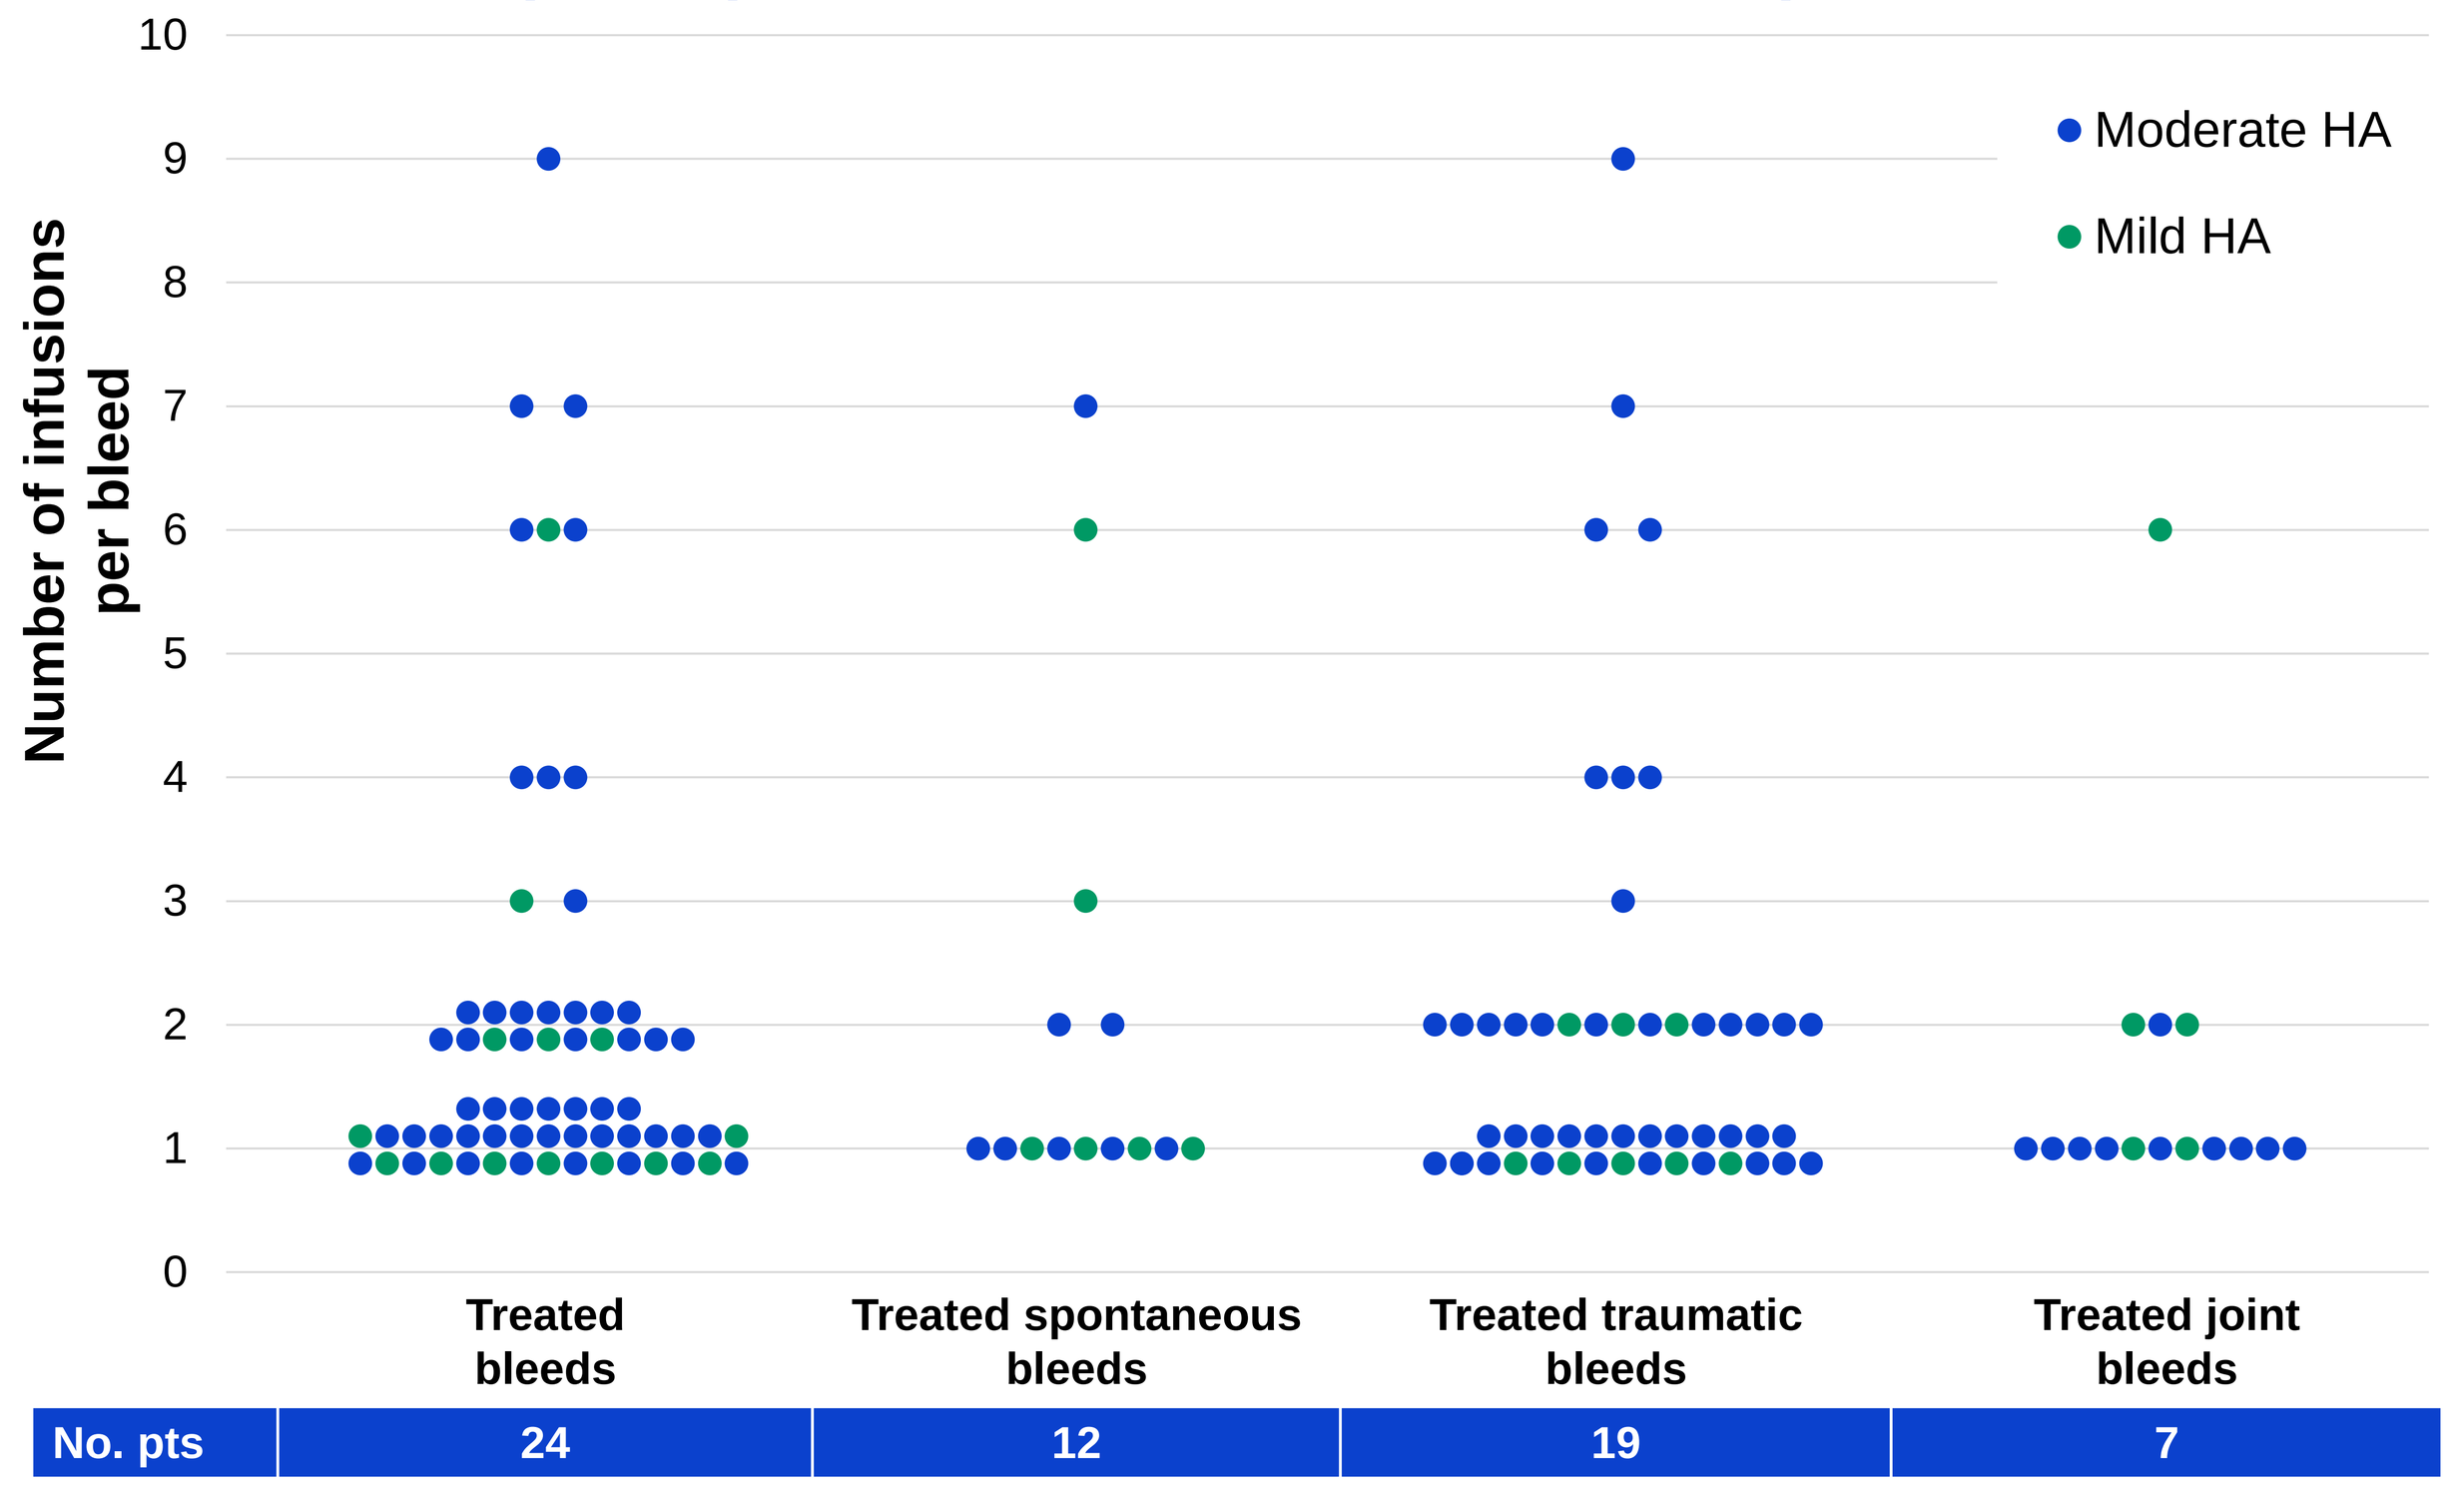

Number of infusions per bleed
10
9
8
7
6
5
4
3
2
1
0
Moderate HA
Mild HA
Treated bleeds
Treated spontaneous bleeds
Treated traumatic bleeds
Treated joint bleeds
No. pts
24
12
19
7

Each point represents one bleeding event. Pt, participant.

Tranexamic acid use for the treatment of breakthrough bleeds

- Eight participants (one with mild HA and seven with moderate HA) received TxA
 - The participant with mild HA received TxA for the treatment of a traumatic rectal bleed caused by ulcerative colitis.
- Of the participants with moderate HA, two received prophylactic TxA for post-operative bleed management (dental procedure and bilateral vasectomy [no bleeding events were reported]) and five received TxA for the treatment of traumatic bleeds (two nose, one eye, one mouth, and one soft tissue)
 - Overall, these participants had 30 medication records, with a total of 93 days of TxA use and a median (range) total duration of TxA use per participant of 10.5 (3–12) days.
- No adverse events were observed with concomitant use of emicizumab and TxA.

Conclusions

- Reported bleeds were mainly traumatic and most resolved after 1–2 doses of replacement FVIII.
- Individual dosing of emicizumab, TxA and FVIII were determined by the investigator and these data may inform breakthrough bleed treatment in people with moderate or mild HA receiving emicizumab.
- No safety concerns were observed with concomitant use of emicizumab and TxA.

Presented at the 2023 American Society of Hematology Annual Meeting | December 9–12, 2023

References

1. Négrier C, et al. Lancet Haematol 2023;10:168–77.
2. Blanchette VS, et al. J Thromb Haemost 2014;12:1935–9.

Acknowledgments

This study was sponsored by F. Hoffmann-La Roche Ltd. Third party medical writing assistance, under the direction of the authors, was provided by Yasmin Bennour, MSc, of Ashfield MedComms, an Inizio company, and was funded by F. Hoffmann-La Roche Ltd.

Disclosures

VJ-Y: consultancy: Grifols, Novo Nordisk, F. Hoffmann-La Roche Ltd, Takeda, Bayer, CSL Behring, Pfizer, BioMarin, Sanofi, Sobi, Spark, Octapharma; research funding: Grifols, Novo Nordisk, F. Hoffmann-La Roche Ltd, Takeda, Bayer, CSL Behring, Pfizer, Sanofi, Sobi, Octapharma; honoraria: Grifols, Novo Nordisk, F. Hoffmann-La Roche Ltd, Takeda, Bayer, CSL Behring, Pfizer, Sanofi, Sobi, Spark, Octapharma; **EL**: employment and equity holder: Genentech, Inc.; **KM**: employment: Genentech, Inc.; **GV**: employment and stock ownership: F. Hoffmann-La Roche Ltd; **AS**: consultancy, honoraria, membership on an entity's Board of Directors or advisory committees, research funding: Novo Nordisk; employment: Indiana Hemophilia and Thrombosis Center; consultancy, honoraria, research funding and speakers bureau: Kedrion Biopharma; honoraria, membership on an entity's Board of Directors or advisory committees, research funding and speakers bureau: Sanofi-Genzyme/Bioverativ, Genentech/Roche; membership on an entity's Board of Directors or advisory committees: Novo Nordisk, Haemophilia Foundation, Hema Biologics, Pfizer; National Hemophilia Program Coordinating Center: Medical Director.